Use of Ex Vivo Precision-Cut Lung Slices as a Screening Tool for Potential Respiratory Toxicity of E-Liquids

H. Raabe[1], M. Aragon[1], D. Sheehan[1], K. Amin[2], M. Oldham[3], W. J. McKinney[3], **K.** M. Lee[3], and H. P. Behrsing[1].

[1]Institute for In Vitro Sciences, Inc., Gaithersburg, MD; [2]University of Minnesota, Minneapolis, MN; and [3]Altria Client Services, Richmond, VA.

ABSTRACT

The Family Smoking Prevention and Tobacco Control Act gave the FDA regulatory authority over next generation tobacco products (NGTP) such as e-vapor products. E-vapor product liquids contain a variety of ingredient combinations that should be assessed for human risk. One human lung-relevant testing platform with reasonable throughput, is human precision-cut lung slices (HuPCLS). HuPCLS are arguably the most complex non-animal model of the lung, retaining native architecture and immune-competent cells over multi-week culture periods. HuPCLS were exposed to three concentrations (0.1%, 0.5%, and 1.2%) of propylene glycol (PG; an E-vapor product constituent) continuously for 16-days. Exposure-effects were evaluated biochemically (WST-8 assay) and histologically (viability assessment of H&E stained slides). Positive control treatments consisted of 10 µM Phortress and 13 µM bleomycin. HuPCLS were fed every day with fresh medium ± treatment and harvested at days 4, 8, and 16. Untreated control (UC) HuPCLS viability was confirmed using protein and adenylate kinase assays. Over 16 days in culture, UC lost 30% viability while WST-8 results indicated no loss over 16 days in culture. Phortress caused severe damage by day 4 and bleomycin by day 8 (histologically & WST-8 viability). Prolonged 1.2% PG exposure diminished WST-8 viability by ~30% at day 16 which agreed with histological results. High osmolality is the suspected mechanism of toxicity. There was no effect histologically or via WST-8 viability for prolonged exposure to 0.1% and 0.5% PG. In summary, PG, a common E-vapor product ingredient, at 1.2% had adverse effects in a human pulmonary model in an exaggerated exposure regimen (prolonged exposures with changes in osmolarity). The HuPCLS platform has huge potential to serve as a screening tool for e-liquid (and other materials of concern) by elucidating potentially relevant, long-term events following NGTP ingredient exposure.

INTRODUCTION

The Family Smoking Prevention and Tobacco Control Act (2009) gives the Food and Drug Administration (FDA) regulatory authority over the manufacture, marketing, and distribution of tobacco products. With the goal of improving public health and as a part of the required information in new premarket tobacco product applications, information must be provided regarding potential health risks associated with a new product.

Respiratory toxicity is specifically listed by the FDA Center for Tobacco Products (CTP) as an important human health effect that can result from using inhalable tobacco products. Substantial research to understand the etiology of chronic pulmonary diseases has led to the development and/or application of a number of in vitro models that are human-relevant. Many available in vitro models for human airways exist and can provide various levels of biological complexity, enabling evaluation of toxicological pathways. However, substantial differences exist between the various existing in vitro models (cell lines, primary cells, and ex vivo tissues) in terms of biological relevance to human organs and capability to model specific events, in addition to cost and performance. For complex biological responses that involve multiple cell types and cell-cell interactions (such as in inflammatory responses), 3-dimensional (3D) tissues are often considered essential.

One of the most physiologically-relevant non-animal models of the lower lung (with reasonable throughput) is precision-cut lung slices (PCLS), offering native lung architecture, including small airways and respiratory parenchyma. These substructures within the human PCLS (HuPCLS) reflect the presence of the many cell types in the human lung, not present in other non-animal models used for toxicity studies. The complement of these cell types in HuPCLS allows for a more realistic interpretation of tissue response to exposures that evoke complex pulmonary responses. We chose to explore use of HuPCLS as a cytotoxicity assay for an ingredient in e-vapor products.

MATERIALS & METHODS

Test System: HuPCLS (donor was a healthy non-smoker), provided by IIAM, Inc.)

Reference materials: Bleomycin Sulfate (13 µM) and Phortress (10 µM)

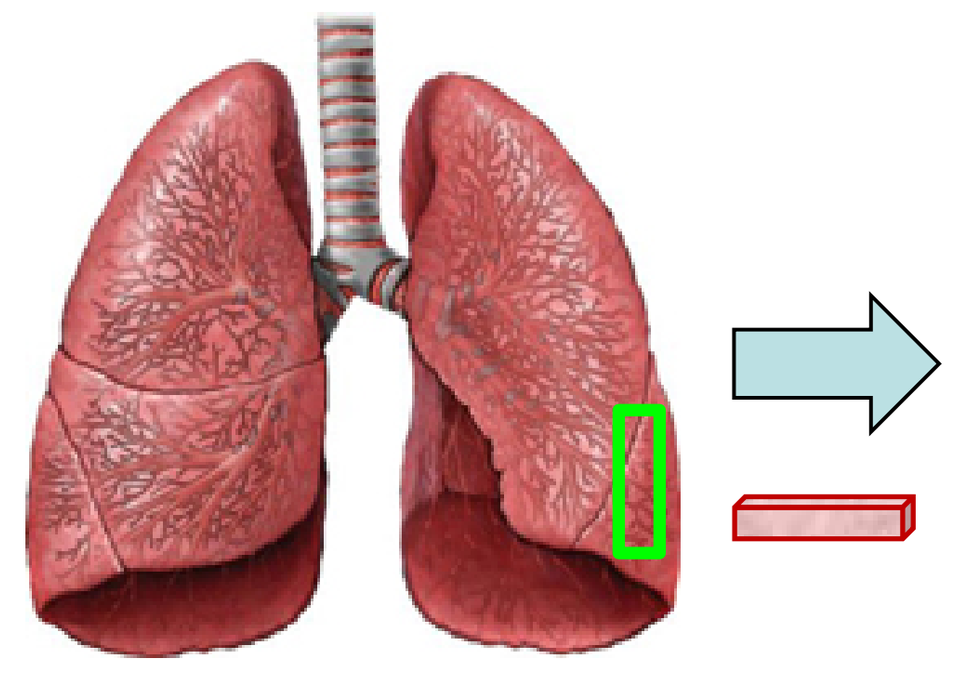

1. Inflate lung tissue and section periphery

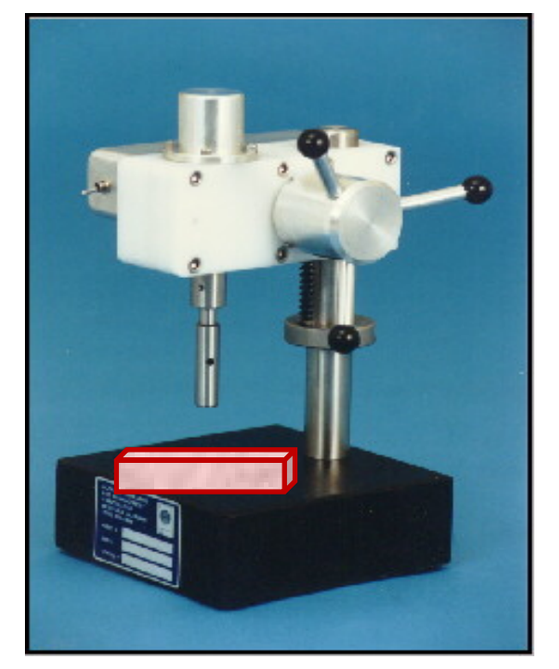

2. Create tissue cores

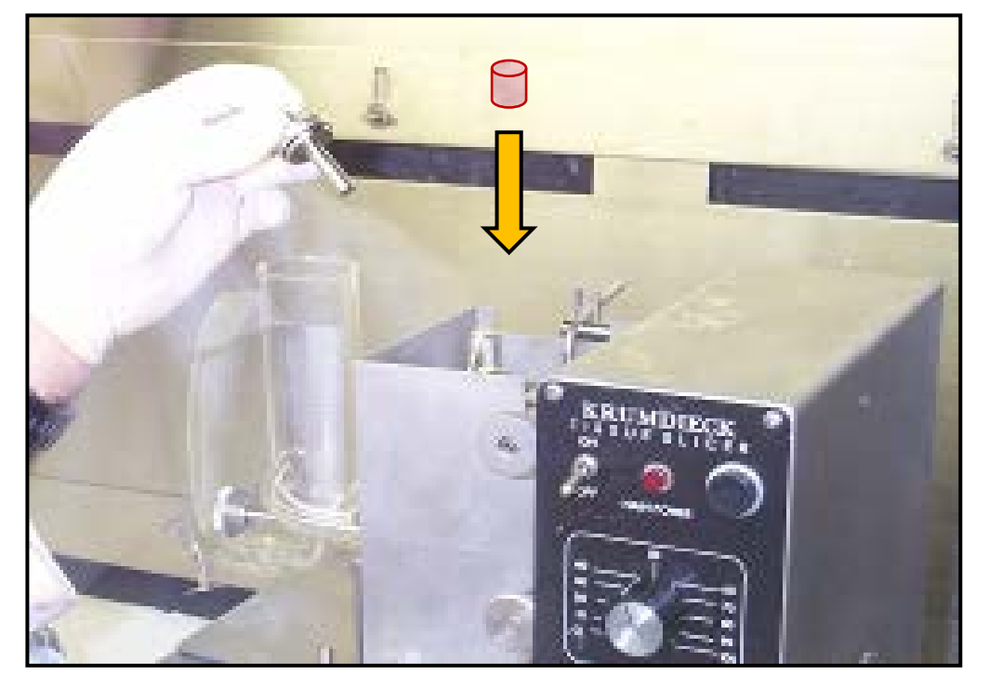

3. Slice cores with slicer

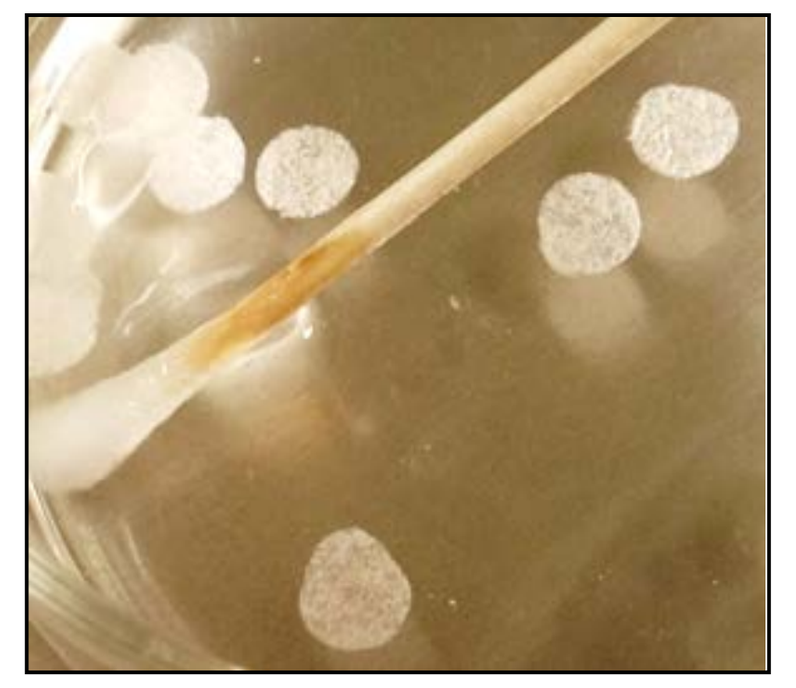

4. Collect slices and place into roller vials

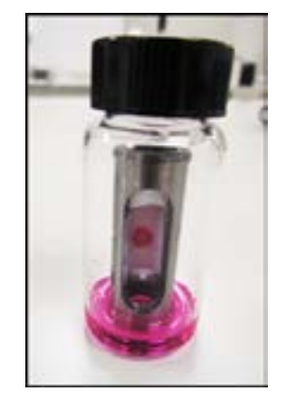

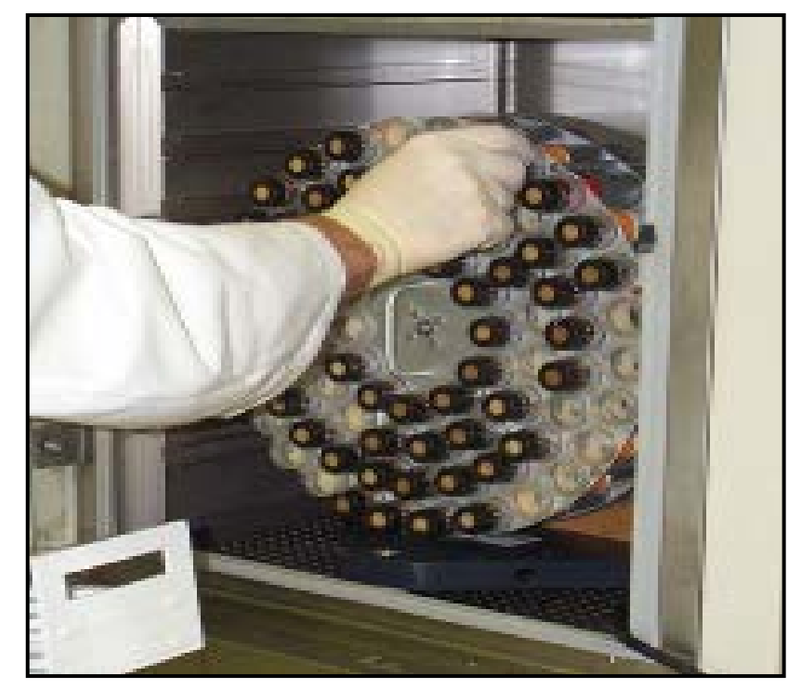

5. Rotate slices at 1-3 rpm in E-199 medium

Procedure:

- After HuPCLS creation and acclimation (4 days), HuPCLS were exposed via media to positive controls (bleomycin or Phortress), the humectant propylene glycol (PG; 0.1%, 0.5%, and 1.2%),or left untreated as the negative control (UC). Slices were fed every day (new media) using each groups respective treatment for a total of 16 days of continuous exposure, post-acclimation. All groups had a minimum of 4 slices per treatment time point.
- Slices were harvested at days 4, 8, and 16. Harvest included HuPCLS undergoing the WST-8 viability assay (Dojindo CCK-8 kit) prior to fixation in neutral buffered formalin. UC groups also had a second set of tissues designated for lysis (in 0.5% triton X-100 in PBS) for total protein and adenylate kinase (AK) content analysis to evaluate the baseline cultures over time.
- Retained treatment media (positive control, PG-containing) was sent to a subcontracting lab (Quality Biological, Inc.) for osmolality measurement.
- Fixed slices were sent for histology processing and H&E staining. Stained HuPCLS sections were then evaluated by a pathologist for hallmark cytomorphological changes, interpreted to assess the overall slice viability and health following exposure.

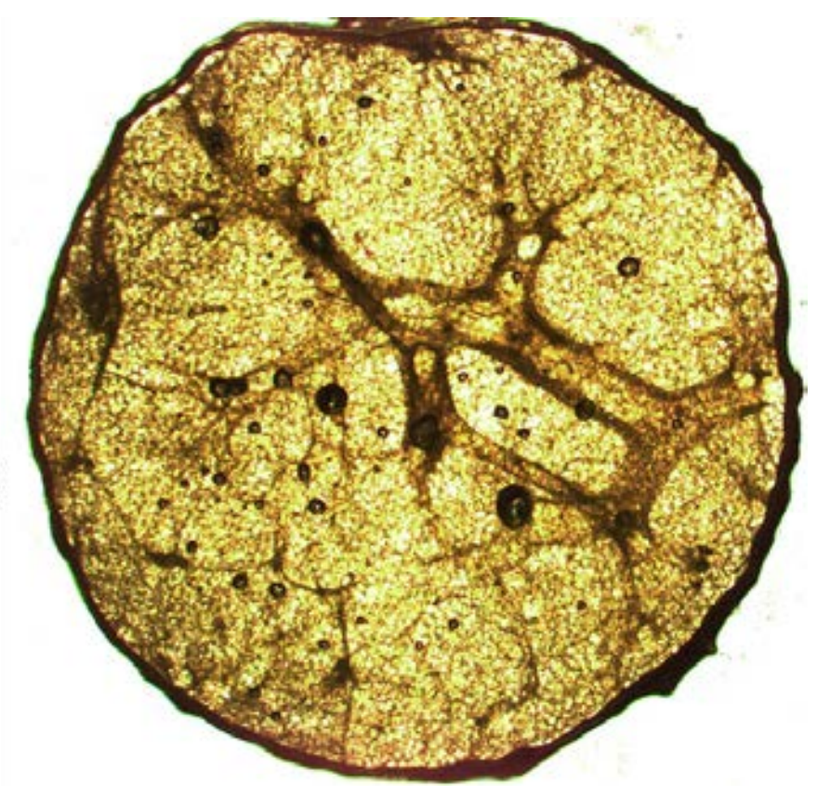

Sample PCLS showing inclusion of airways and respiratory parenchyma

RESULTS

Culture Longevity of Untreated Control (UC) HuPCLS

Table 1: Markers of UC HuPCLS over time in Culture

Assay:	WST-8 Viability[1]	AK Content[2]	Protein Content[2]	Histology Viability[3]
	Group Averages & Standard Deviation (SD)			
Day	Abs (450nm) SD	RLU /PCLS	mg /PCLS	Score (0-4) SD
0	1.5 ± 0.4	54797	0.11	0.0 ± 0.0
4	1.7 ± 0.2	73832	0.12	0.0 ± 0.0
8	1.4 ± 0.1	77850	0.10	0.7 ± 0.6
16	1.4 ± 0.3	131600	0.16	1.3 ± 0.7

[1] Average derived from 4-12 PCLS/time point

[2] Average derived from 4 PCLS, harvested in pairs

[3] Average derived from 4-7 PCLS/time point

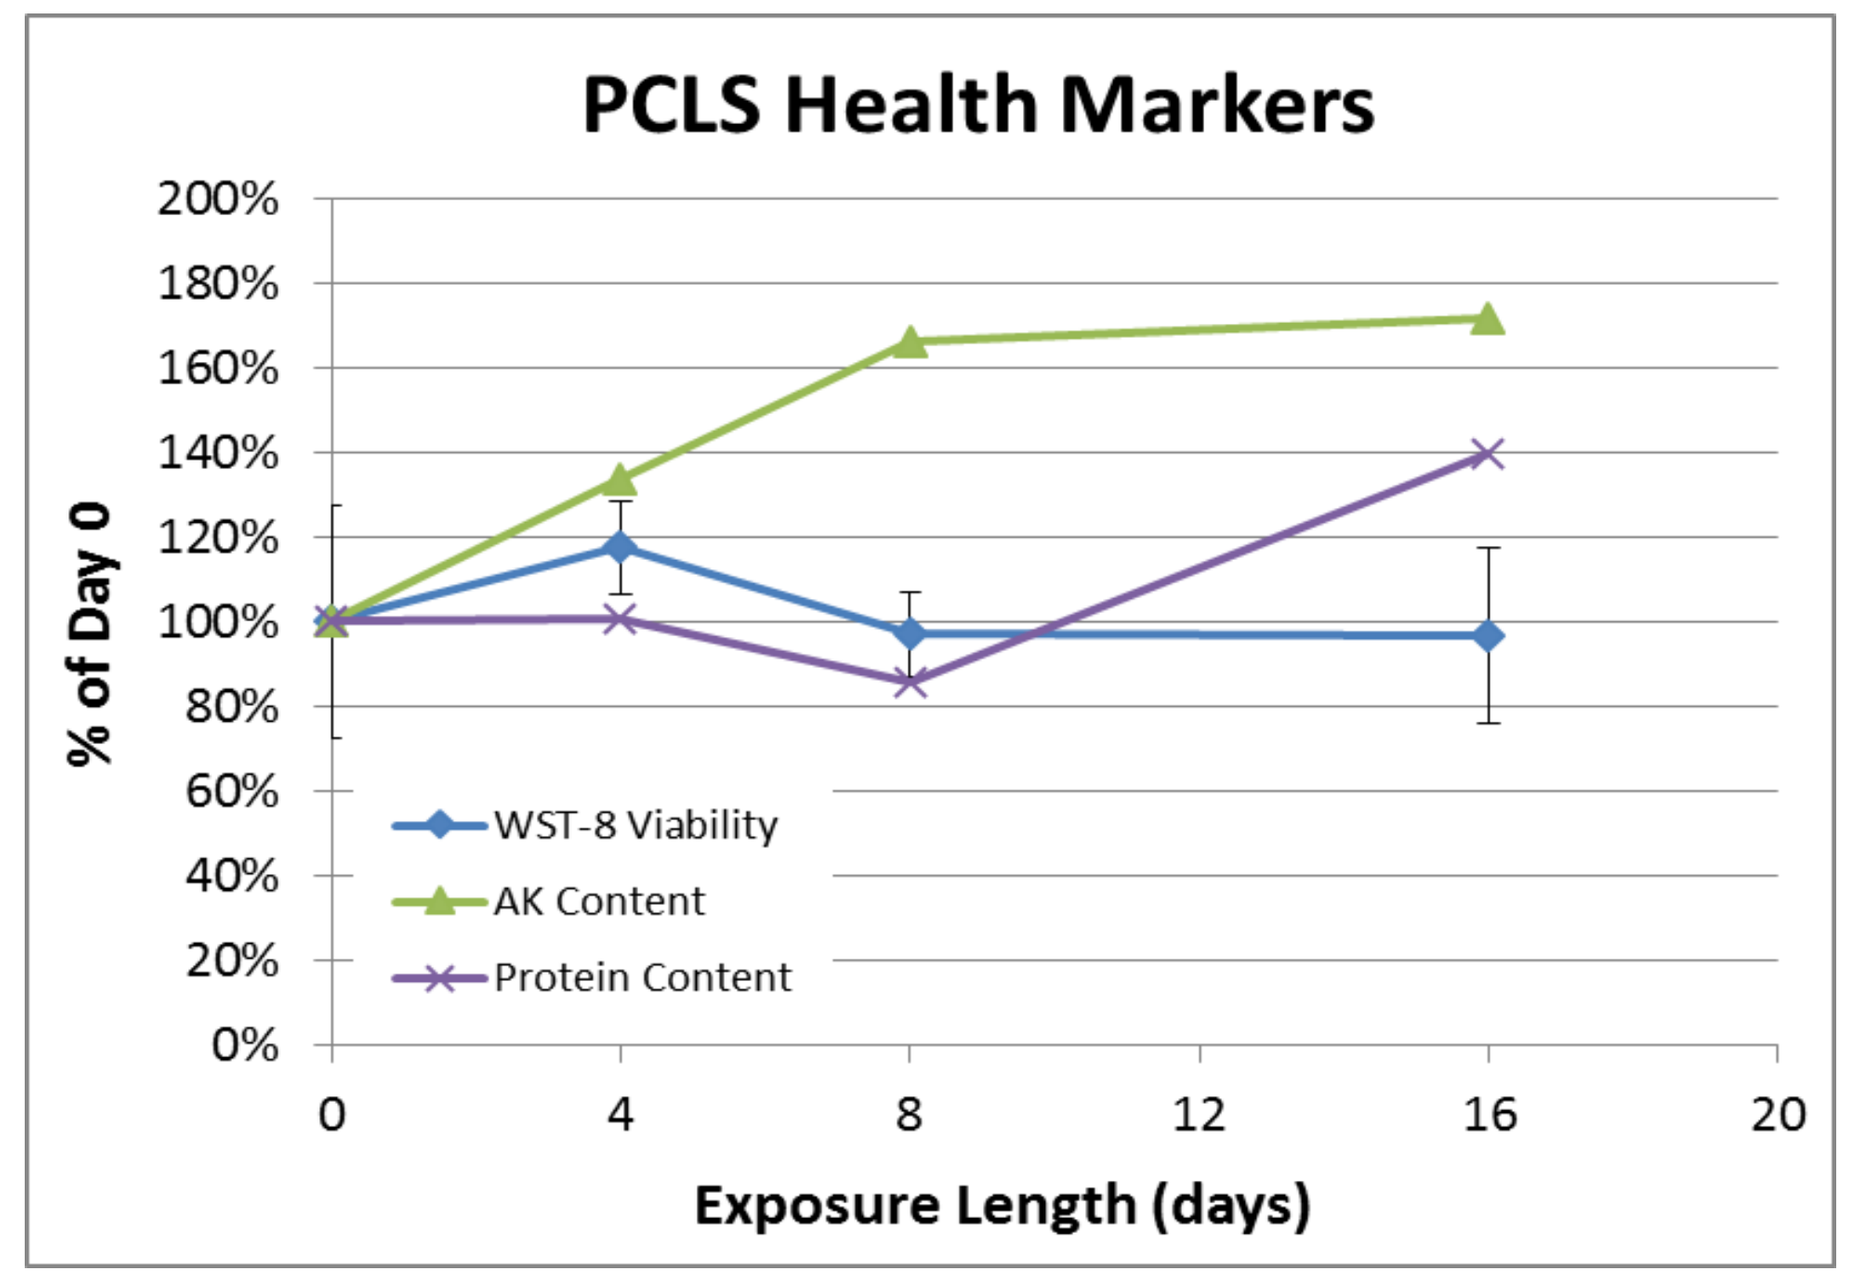
PCLS Health Markers
200%
180%
160%
140%
120%
100%
80%
60%
40%
20%
0%
% of Day 0
WST-8 Viability
AK Content
Protein Content
0
4
8
12
16
20
Exposure Length (days)

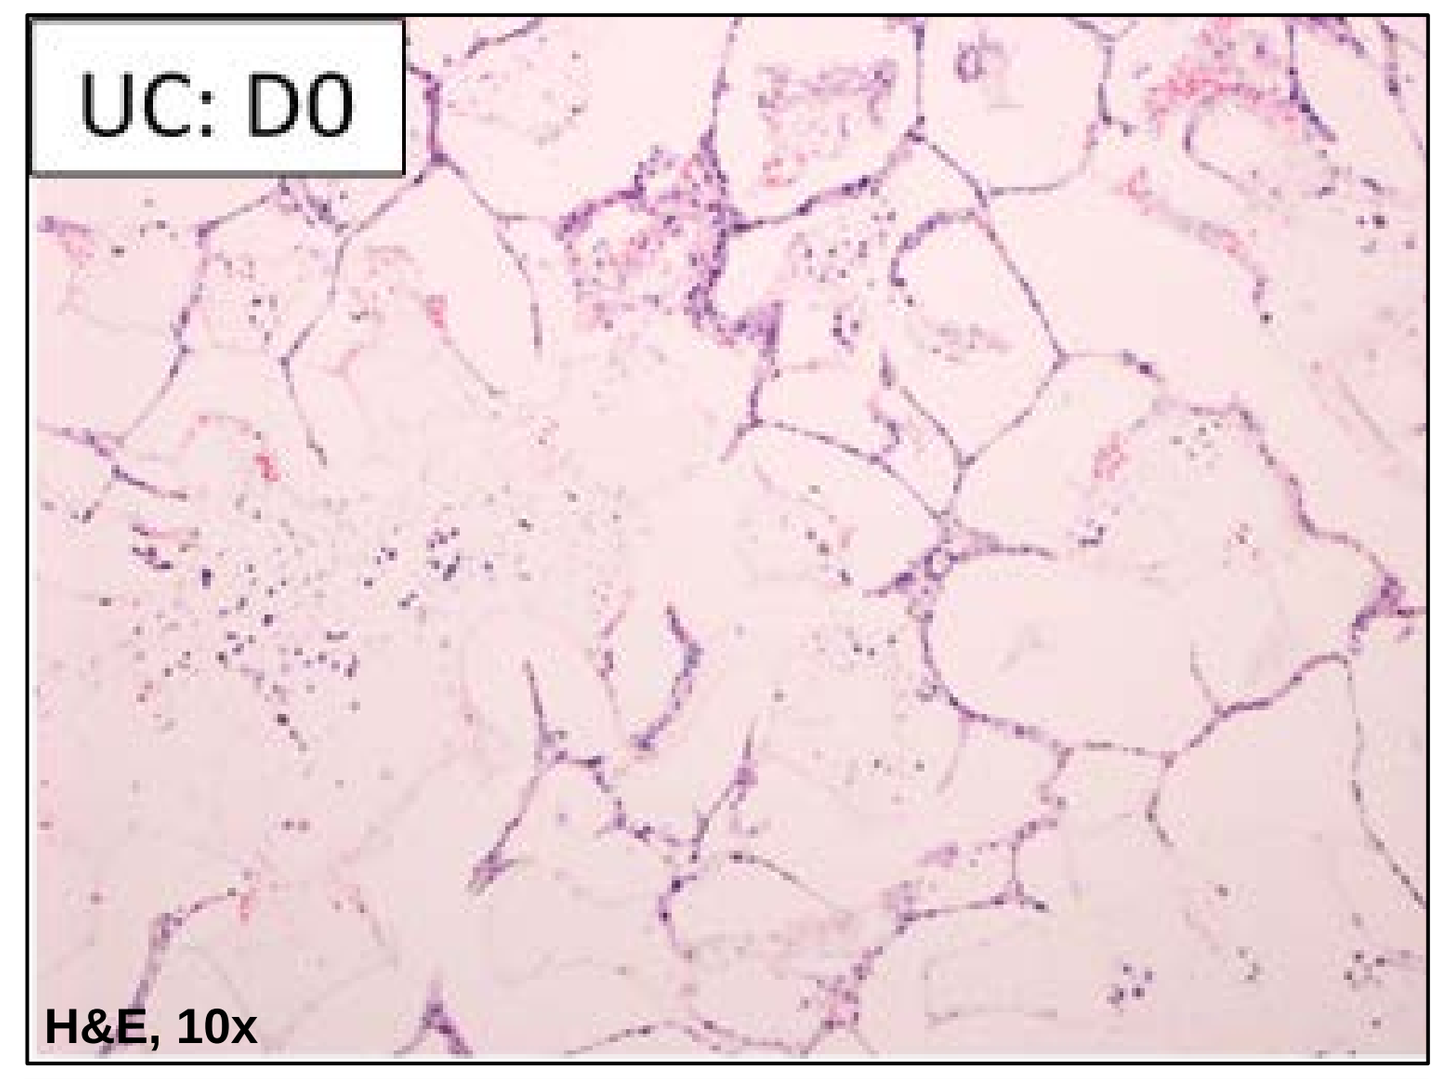
UC: D0
H&E, 10x

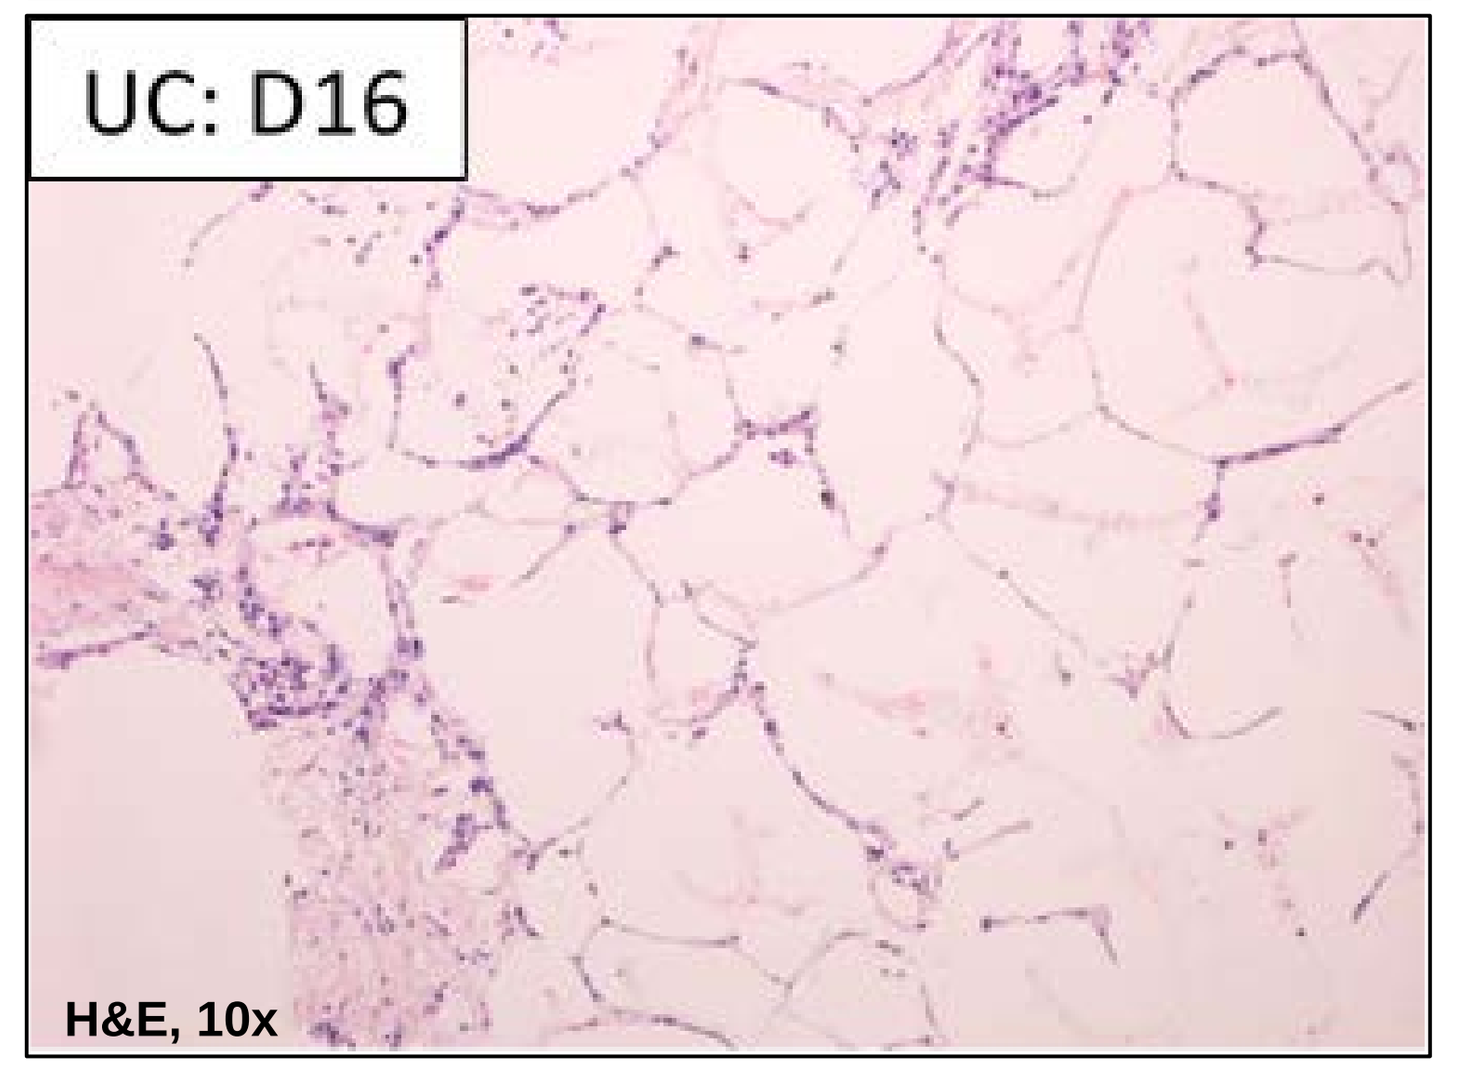
UC: D16
H&E, 10x

- Multiple endpoints suggests HuPCLS are viable over the 16-day culture period.
- The biochemical data (WST-8 viability and tissue protein content) showed minimal changes (within 20%) up to 8 days and a modest increase in protein on day 16. Increase of tissue AK activity over time may be a reflection of slices responding to culture conditions.
- Evaluation of H&E slides where cytomorphological changes are interpreted to assess changes in viability indicate a minor loss of viability during the 16 days in culture.
- Results suggest that under the experimental conditions, the HuPCLS are fit for cytotoxicity testing up to 16 days.

RESULTS cont.

Viability & Osmolality

WST-8 Results:

- Positive Controls elicited tissue damage as expected, with Phortress causing severe damage (3% of UC viability remaining) by day 4 harvest. Bleomycin-induced damage progressively worsens , with only 7% of UC viability remaining by day 16.
- PG groups did not cause any statistically significant (2-tailed T-test, unequal variance) loss of WST-8 viability until day 16. Of the three PG concentrations tested, only 1.2% measured lower than the UC group at day 16.

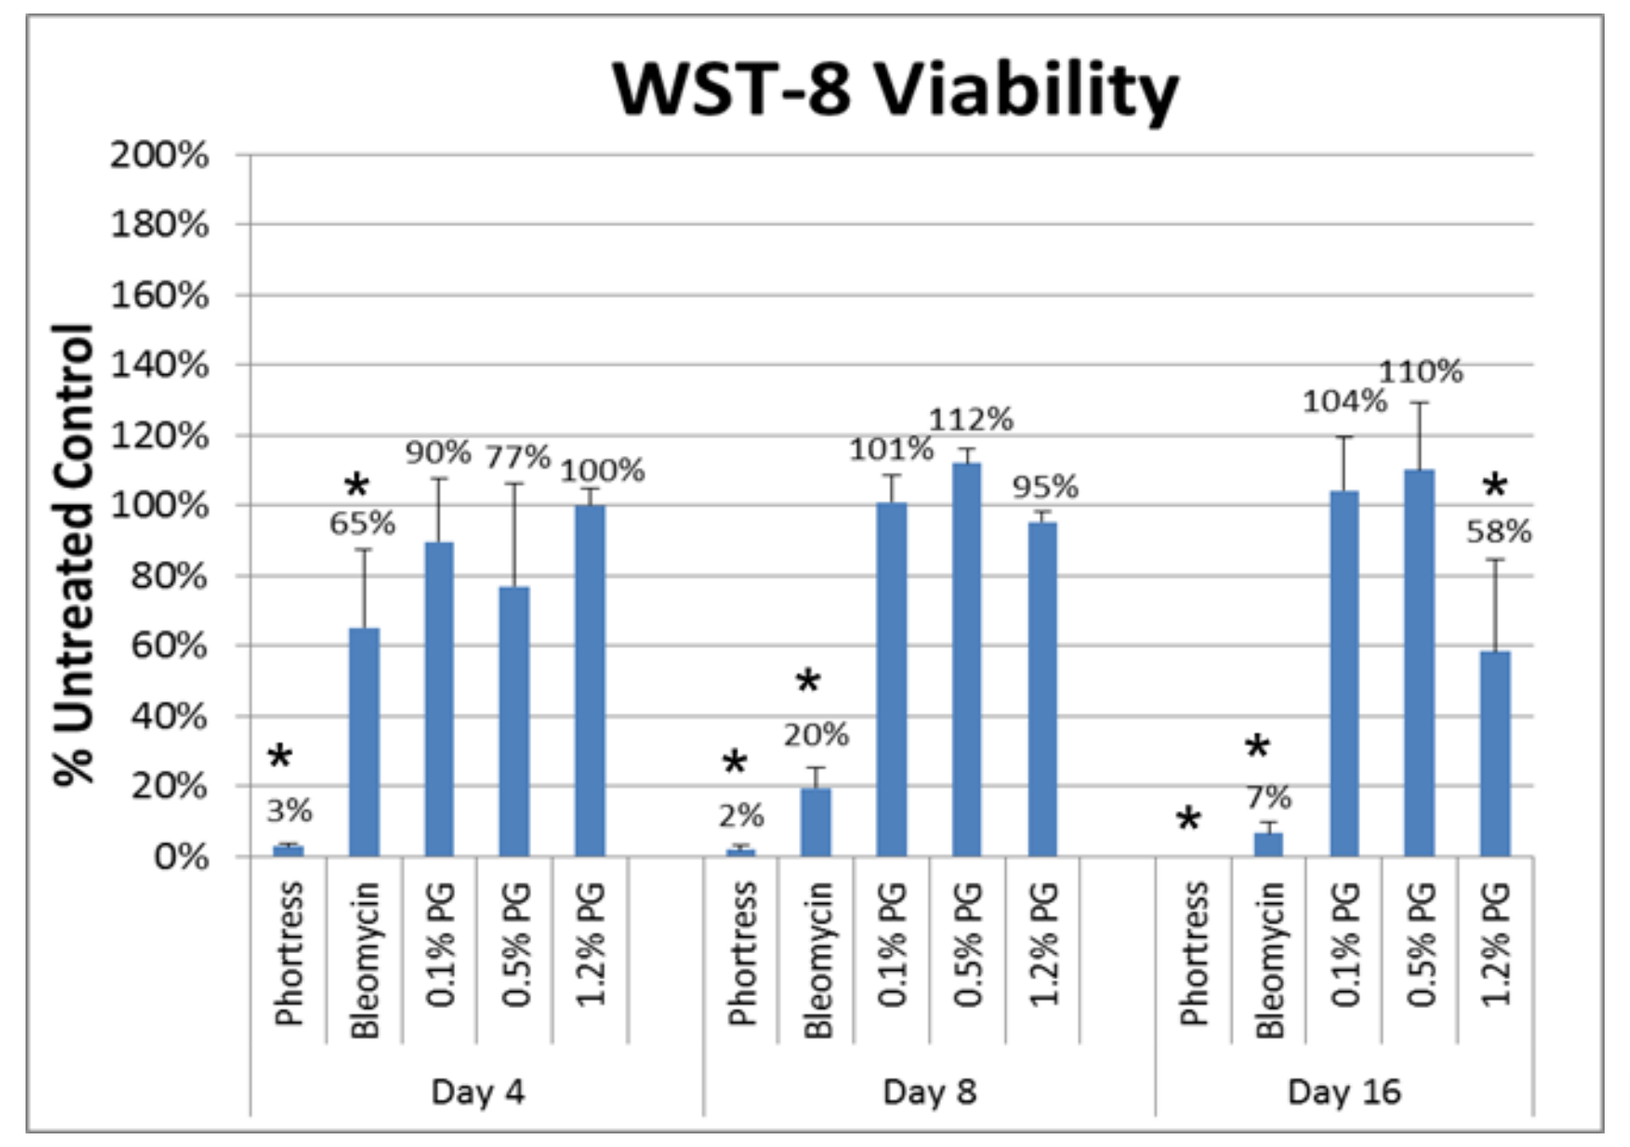
WST-8 Viability
200%
180%
160%
140%
120%
100%
80%
60%
40%
20%
0%
% Untreated Control
*
3%
*
65%
90%
77%
100%
*
2%
*
20%
101%
112%
95%
*
*
7%
104%
110%
*
58%
Phortress
Bleomycin
0.1% PG
0.5% PG
1.2% PG
Day 4
Day 8
Day 16

* *Significantly lower than untreated control (p<0.05)*

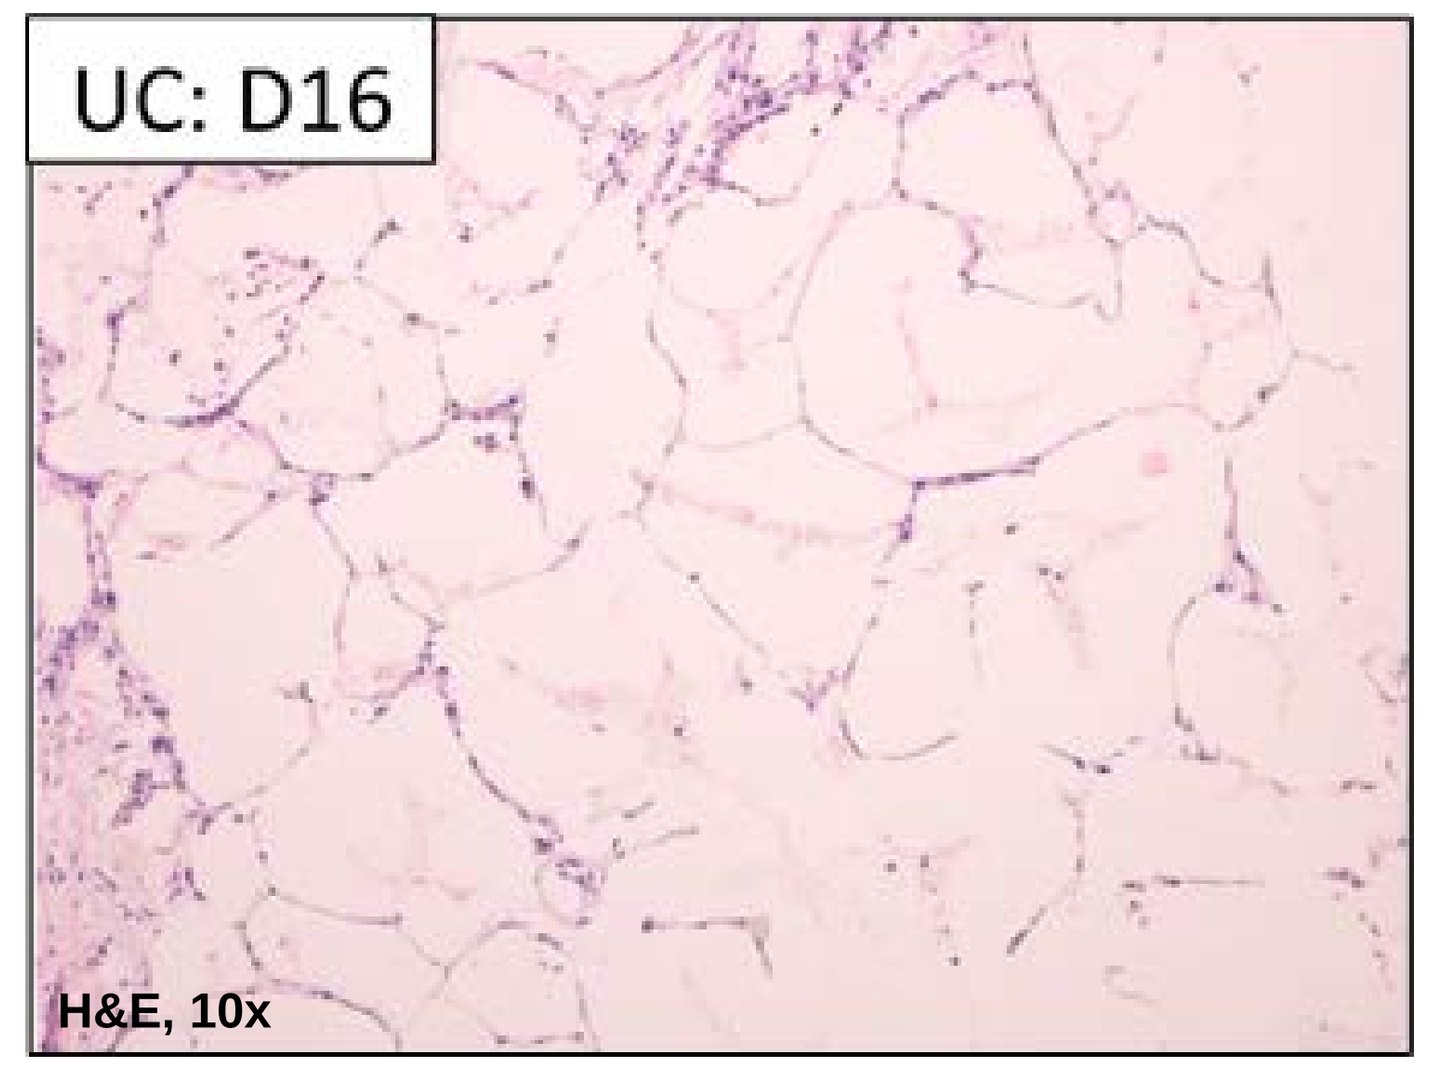
UC: D16
H&E, 10x

Table 2: Average (AVG) viability score (0-4) derived from H&E slides

Treatment	Day 0[1] AVG ± SD	Day 4 AVG ± SD	Day 8 AVG ± SD	Day 16 AVG ± SD
Untreated Control	0.0 ± 0.0	0.0 ± 0.0	0.7 ± 0.6	1.3 ± 0.7
10 µM Phortress		2.0 ± 0.0	4.0 ± 0.0	4.0 ± 0.0
13 µM Bleomycin		1.0 ± 0.0	3.0 ± 0.0	4.0 ± 0.0
0.1 % PG		0.8 ± 0.5	1.5 ± 0.6	1.3 ± 0.6
0.5 % PG		1.8 ± 1.0	1.0 ± 0.0	1.0 ± 0.0
1.2 % PG		0.0 ± 0.0	1.0 ± 0.0	1.5 ± 0.6

0-4 viability score is derived from % viability, using the scoring conversion as follows: "0" = 81-100%; "1" = 61-80%; "2" = 41-60%; "3" = 21-40%; "4" = 0-20%.

[1] all treatment groups share the post-acclimation Day 0 UC group as a pre-exposure control

Histology Results:

- Positive Controls were confirmed as causing damage, with moderate-severe loss of viability assessed by day 4 and 8, with Phortress- and bleomycin-exposure, respectively
- All PG groups appeared similar to the UC group at all time points

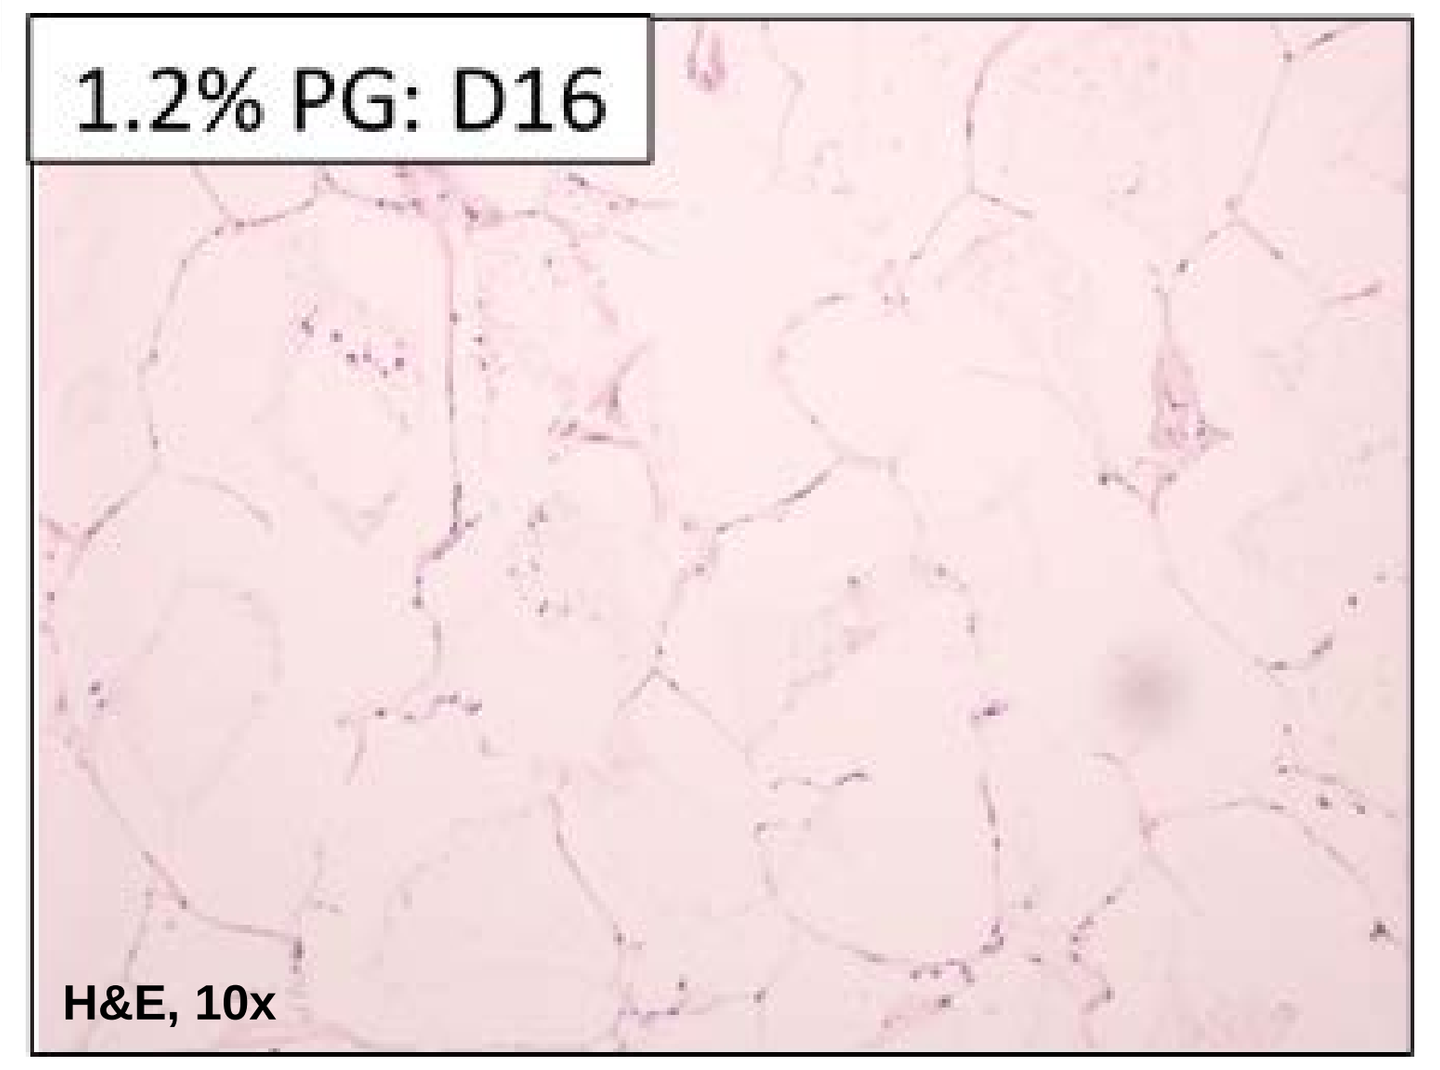
1.2% PG: D16
H&E, 10x

Osmolality Results:

- All treatment media were tested for osmolality and compared to the accepted human physiological range (275-295 mOsm/kg)
- Of all exposure media, only the 0.5% and 1.2% PG groups had measured values higher than the normal human range. Of these, only 1.2% PG caused notable change in the WST-8 viability assay.

Table 3: Osmolality Readings

Sample	mOsm/kg A	mOsm/kg B	AVG
Culture Medium (CM)	289	287	288
13 µM Bleomycin in CM	286	288	287
10 µM Phortress in CM	288	288	288
0.1% PG in CM	299	300	300
0.5% PG in CM	363	362	363
1.2% PG in CM	453	453	453

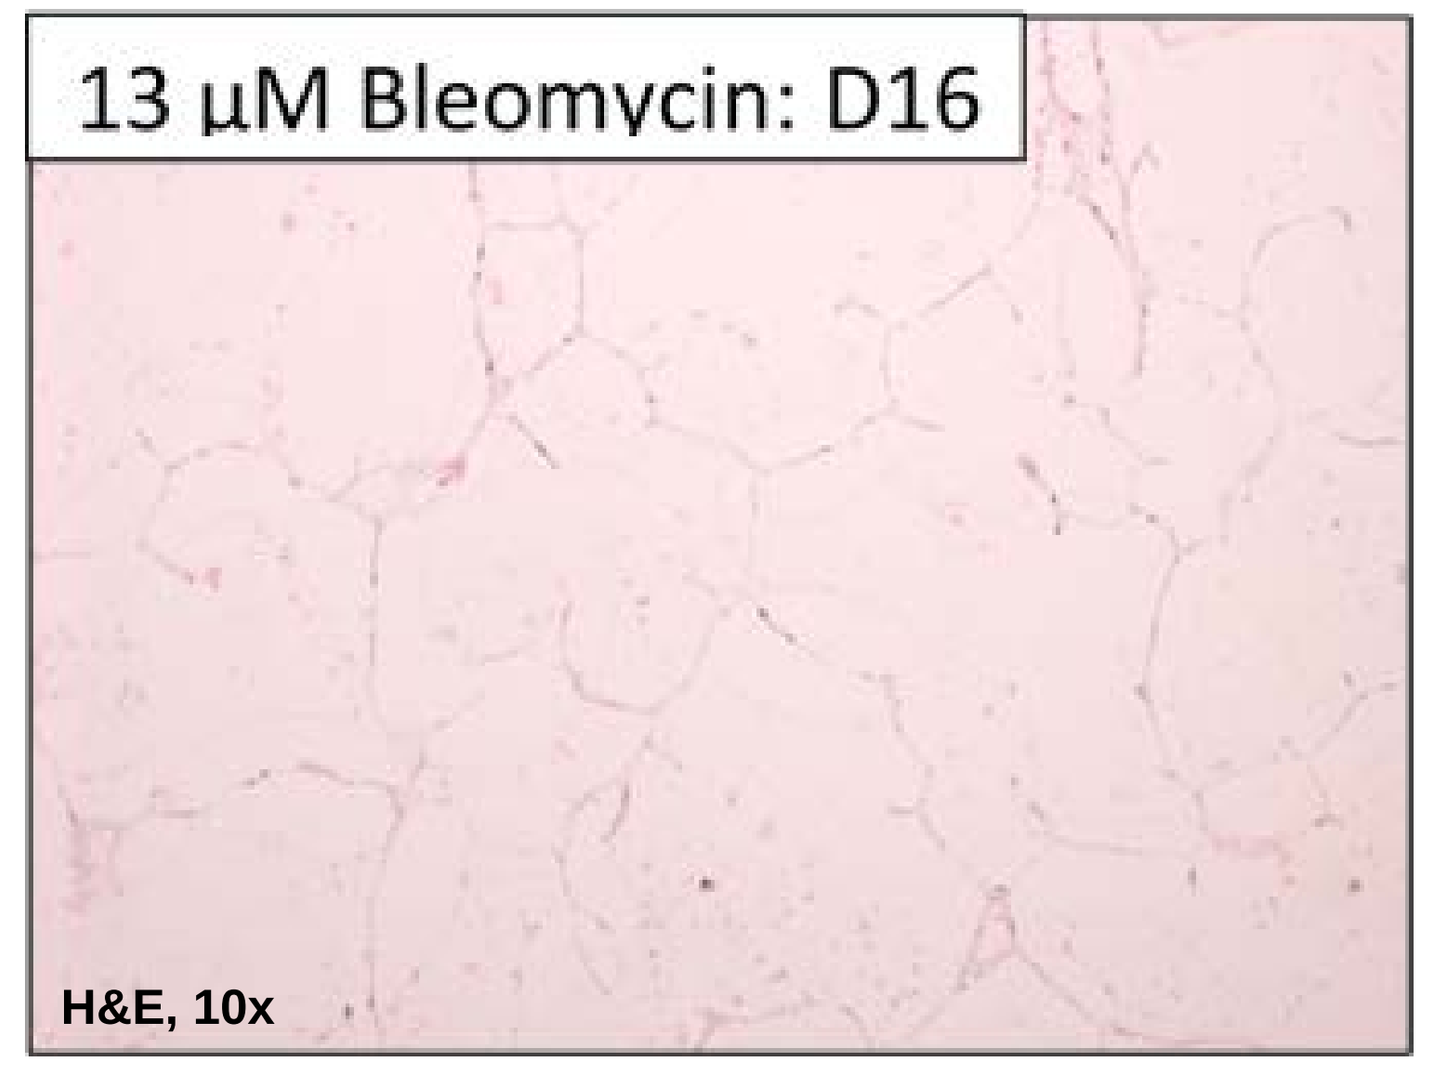
13 µM Bleomycin: D16
H&E, 10x

CONCLUSIONS

- For 16 days post-acclimation, HuPCLS exhibited characteristic retention of overall health and viability, as measured using the WST-8 viability assay, tissue content of AK, and protein, and finally through the histological interpretation of H&E stained slides.
- Combining the biochemical and histological assessment of HuPCLS viability allows for a more thorough assessment of treatment impact on human lung tissue, with differences in viability measures attributable to the inherent assay variability.
- The reference materials (PC) performed as expected. The high osmolality of the exposure media containing the 1.2% PG is hypothesized to be the cause of the significant loss of WST-8 viability – it is expected that a longer culture would have shown this effect histologically.
- Development and Validation of HuPCLS as a cytotoxicity assay will require a significant amount of additional replicate experiments, testing of additional ingredients and also an expansion of the battery of toxicity endpoints.

REFERENCES

- Behrsing, H. P., et al. (2013). "In vitro exposure of precision-cut lung slices to 2-(4-amino-3-methylphenyl)-5-fluorobenzothiazole lysylamide dihydrochloride (NSC 710305, Phortress) increases inflammatory cytokine content and tissue damage." Toxicol Sci 131(2): 470-479.
- Tyson, C. A., K. Amin, et al. (2005). "Comparison of BCNU and SarCNU Toxicity in Long-Term Cultures of Precision-cut Lung Slices." The Toxicologist 84: 240.