Comparison of *In Vitro* Toxicity of E-Vapor Condensates to Literature Data for Cigarette Smoke Condensates

Altria
Altria Client Services

RAMBØLL

AUTHORS

MM Moore, Ramboll; BG Coffa, U Doshi, KM Lee, Altria Client Services LLC

ABSTRACT

The US Food and Drug Administration Draft Guidance (2016) "Premarket Tobacco Product Applications for Electronic Nicotine Delivery Systems" recommends toxicological evaluations, including assessing *in vitro* genotoxicity and cytotoxicity, for new electronic nicotine delivery systems (ENDS). The draft guidance also recommends that the toxicity of the ENDS be compared with that of other tobacco products, such as combustible cigarettes. In the current analysis, we compared the genotoxic/cytotoxic potency (biological response per mass quantity) of condensates from selected e-vapor ENDS products marketed as MarkTen® (MT) with the condensates from combustible cigarettes. For the e-vapor products, the aerosol condensates were collected from MT products with a total of 7 formulations (one carrier [propylene glycol, glycerine, plus 5% nicotine] and 6 e-liquids [carrier plus flavor mixtures]). The e-vapor condensates were tested in 2 genotoxicity assays (Ames Test and *in vitro* micronucleus assay [MN] using TK6 cells) and the neutral red uptake (NRU) cytotoxicity assay according to the corresponding OECD guidelines. For the Ames Test and the NRU assay, literature data describing the range of responses for cigarettes were available for comparisons. Several published studies using a variety of cigarettes and cell types provide an overall assessment of cigarettes in the MN assay. All of the cigarette condensates were genotoxic in the Ames and MN assays and cytotoxic in the NRU assay. The e-vapor condensates were not cytotoxic (NRU assay viability >80%) and not genotoxic in the Ames Test or the MN assay at any of the tested concentrations.

GOALS

- To review literature and summarize the genotoxicity of cigarette smoke condensates (CSCs) (Ames Test and *in vitro* MN assay) and cytotoxicity (NRU) for a broad range of combustible cigarettes
- To assess the potential genotoxicity/cytotoxicity of condensates from selected e-cigarettes (one carrier [propylene glycol, glycerine, plus 5% nicotine] and 6 e-liquids [carrier plus flavor mixtures]) from MT e-vapor products
- To compare the genotoxicity/cytotoxicity of the MT e-condensates and selected CSCs as reported in literature

E-CONDENSATE COLLECTION FROM ENDS (MT)

Aerosols were generated from the MT device using a modified Health Canada intense regimen (55 mL puff volume, 30 sec frequency, 5-sec duration and a square puff wave profile). To complete a replicate, 140 puffs were collected from each of two cartridges. Total particulate matter (TPM) was collected on a non-conditioned 55 mm Cambridge filter pad (CFP) followed by impinger filled with 30 mL USP-grade ethanol cooled in an ice water bath.

The CFP was extracted with impinger contents and then filtered through a sterile cheesecloth to produce the condensates. Eight collections were pooled (from a total of 16 cartridges) into a single composite condensate sample (the target concentration of 37–50 mg/mL in ethanol).

Figure 1. Ames mutagenic potencies (TA98 with S9) for combustible cigarette condensates (>300 different cigarettes, combining data from DeMarini et al 2008; Gaworski et al 2011a,b; Steele et al 1995)

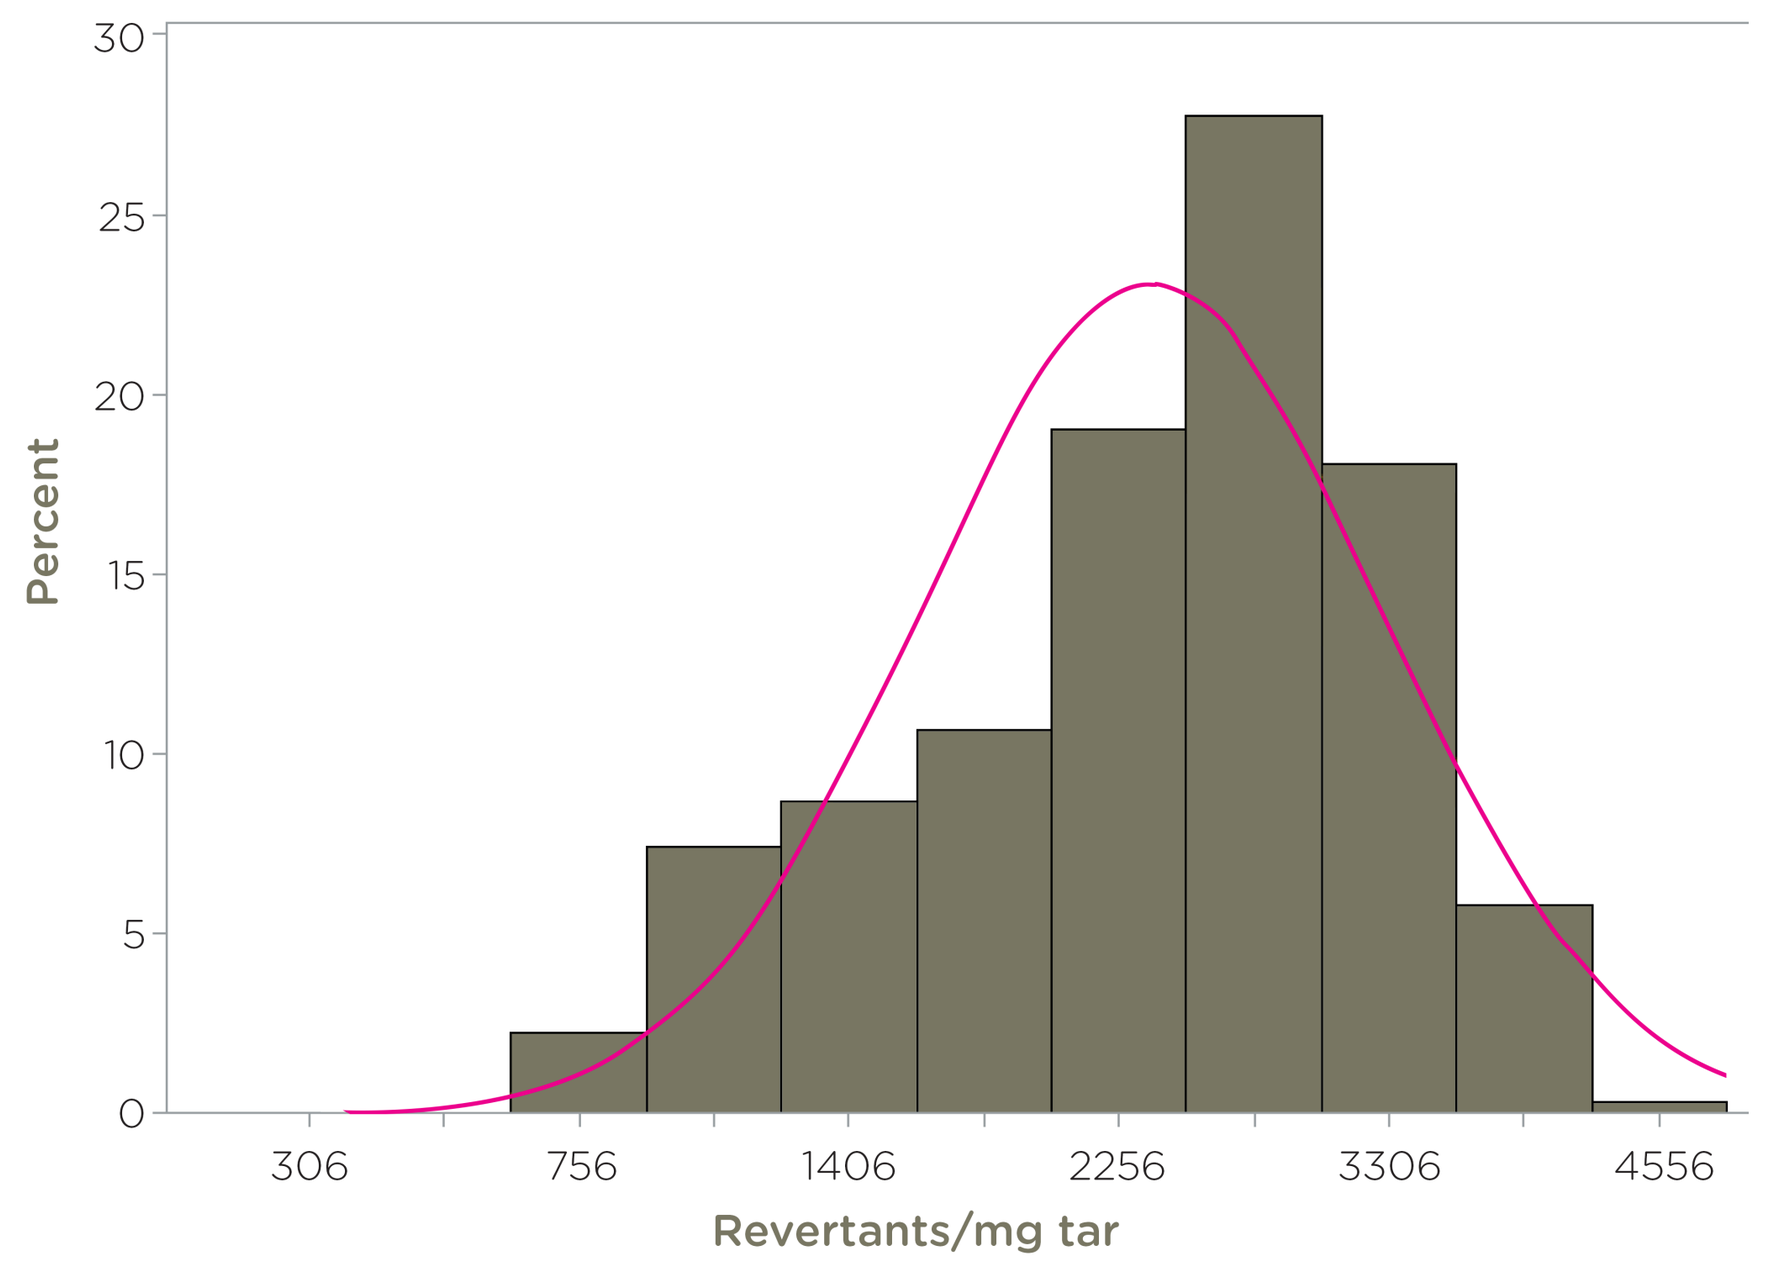
Percent
Revertants/mg tar
306
756
1406
2256
3306
4556

AMES TEST

The Ames mutagenic potencies for a range of combustible cigarette condensates (CSCs) were obtained from literature (DeMarini et al 2008; Steele et al 1995; Gaworski et al 2011a,b). The CSCs were generated under the ISO smoking protocol (35 mL puff volume, 60 sec frequency, 2 sec duration) and the potency values were obtained based on the linear model (as recommended by Bernstein et al 1982). Strains TA98 and TA100 are responsive to CSC, with TA98 being the most responsive. Thus, we used data from TA98 to represent the mutagenicity of the CSCs. Using the approach reported by Steele et al (1995) to graphically present the distribution of potencies in TA98 for the 73 US marketed brand cigarettes, the potency data from Steele et al were combined with that from Gaworski et al (2011a,b) and DeMarini et al (2008). This approach provided Ames data from >300 combustible cigarettes (Figure 1).

The e-condensates were evaluated using 5 strains, according to OECD TG471 (OECD 1997) using the preincubation method with and without S9. The top concentrations of the e-condensate ranged from 2,820 to 4,807 µg/plate TPM (total particulate matter).

For all e-condensates, no positive trend was observed in the 5 strains (data shown only for TA98; see Figure 2). To compare the mutagenic potential of CSCs and e-condensates, the dose-response data (from Table 2 of DeMarini et al 2008) are shown with the dose-response data for the MT e-condensates (Figure 2).

The CSC from most of the cigarettes are mutagenic at concentrations of 100 µg/plate and the linear portion of the dose-response curve occurs between 0 and approximately 400 µg/plate TPM. None of the e-condensates induced any increase in the number of revertants even at much higher TPM levels.

IN VITRO MICRONUCLEUS (MN) ASSAY

The literature search did not identify any publications in which TK6 cells were used to test CSCs for their potential to induce MN. However, publications reporting *in vitro* MN results for CSCs using OECD TG487 recommended cell lines, such as CHO, V79 and L5178Y indicate that CSCs are all MN positive (genotoxic). In lieu of TK6 cell MN literature data for CSCs, we used the 2017 ALCS's CORESTA proficiency trial data where 3R4F reference CSC was evaluated in the *in vitro* MN assay.

For the e-condensates, the *in vitro* MN assay was conducted according to OECD TG487 (OECD 2016) using 3 different treatment conditions, a short 4-hr treatment without S9, a short 4-hr treatment with S9 and a long 27-hr treatment without S9. The top concentration for all e-condensates was limited by the amount of solvent (1% ethanol) that could be added to the cell culture. The actual top concentrations ranged from 282 to 481 µg/mL TPM.

Cytotoxicity was assessed using relative population doubling (RPD). For the 4-hr treatments, cells were harvested ~40 hr after the treatment, and for the 27-hr treatment, cells were harvested immediately. At least 1,000 cells were scored for each of the duplicate cultures, with 2,000 cells per concentration evaluated.

Figures 3–5 demonstrate the significant difference between the 3R4F CSC and e-condensates with regard to their cytotoxicity and genotoxicity in TK6 cells.

For 3R4F CSC, the concentrations tested (up to 120 µg/mL) give a clear dose-related increase in cytotoxicity of 55% (or 45% RPD), while e-condensates show little to no difference compared to the concurrent vehicle control for concentrations up to ~480 µg/mL. Furthermore, a direct comparison between the 3R4F CSC and e-condensates (Figures 3–5) shows that while 3R4F CSC indicates a dose-related increase in the %MN under all 3 test conditions, the e-condensates show no dose-related increase.

NEUTRAL RED UPTAKE ASSAY

The cytotoxicity (based on EC_{50} in the NRU using BALB/c 3T3 cells) of CSCs from a range of cigarette products were evaluated using data from Gaworski et al (2011a,b) (Figure 6).

The NRU cytotoxicity assessment of e-condensates was conducted using BALB/c 3T3 cells according to the recommendations in OECD TG129 (OECD 2010). Cells were seeded and treated with either solvent or 1 of 8 concentrations of e-condensates. The top concentration was determined by the amount of solvent (0.5% ethanol) that could be delivered to the cells.

The top concentrations ranged from 141 to 240 µg/mL TPM. After a 48-hr incubation, the cells were washed with PBS and exposed to neutral red dye. The plates were read using a spectrophotometer at 540 nm ± 10 nm. For e-condensates, none of the test concentrations reduced the cell density by 50%, and therefore EC_{50} was not calculated.

Figure 6. The distribution of the EC_{50} responses observed by Gaworski et al (2011a,b)

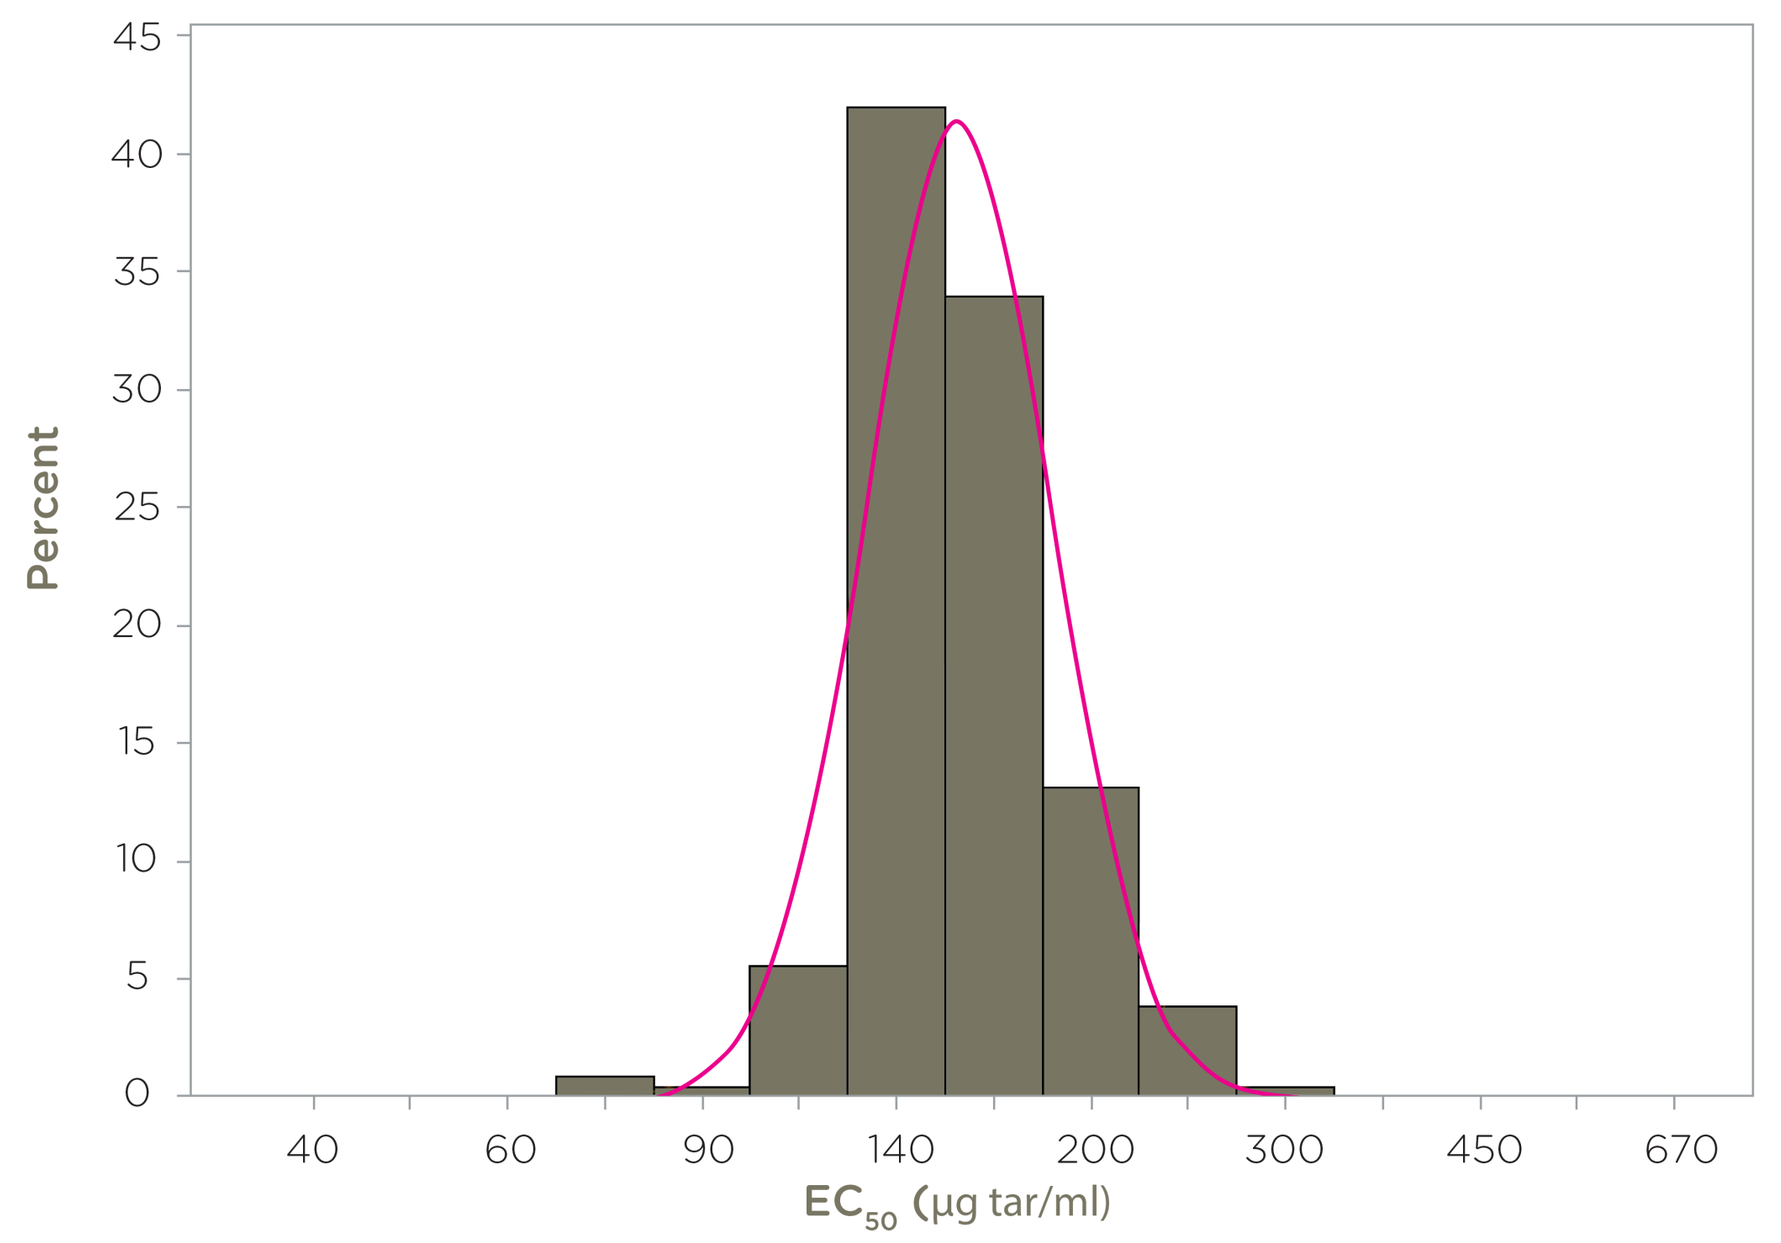
Percent
EC50 (µg tar/ml)
40
60
90
140
200
300
450
670

All the CSCs from Gaworski et al (2011a,b) were cytotoxic (Figure 6). In contrast, the e-condensates were not cytotoxic even when tested to the maximum concentration. None of the e-condensates approached the ISO 10993-5 definition of cytotoxic chemicals (<70% cell viability when compared to the concurrent solvent control) (ISO 2009) (Figure 7).

This poster may be accessed at www.altria.com/ALCS-Science.

CONCLUSIONS

- Combustible cigarette condensates are mutagenic and genotoxic, inducing positive responses in the Ames Test and the *in vitro* MN assay.
- Combustible cigarette condensates are cytotoxic to cells in culture and EC_{50}s are readily calculated.
- Condensates from e-cigarettes were not mutagenic nor genotoxic in the Ames Test and the *in vitro* MN assay when evaluated to the top concentration feasible based on the amount of solvent added to the test cultures.
- E-condensates were not cytotoxic (NRU assay viability >80%).
- All of the comparisons (Ames Test, the *in vitro* MN assay and the NRU assay) show clear differences in biological activity between combustible cigarettes and e-cigarettes.

REFERENCES

Bernstein L, et al 1982. Mutation Research, 97(4): 267–281.

DeMarini DM et al 2008. Mutation Research, 650(1): 15–29.

Gaworski CL et al 2011a. Inhalation Toxicology, 23(S1): 1–12.

Gaworski CL et al 2011b. Inhalation Toxicology, 23(S1): 172–183.

ISO. 2009. ISO: 10993-5 Biological evaluation of medical devices Part 5: Tests for *in vitro* cytotoxicity. International Organization for Standardization.

Steele RH et al 1995. Mutation Research, 342(3-4): 179–190.

USFDA/CTP. 2016. Premarket tobacco product applications for electronic nicotine delivery systems: Guidance for industry. United States Department of Health and Human Services, Food and Drug Administration, Center for Tobacco Products.

Figure 2. Ames Test TA98 with S9. Dose response for 10 combustible cigarettes (dotted lines) and for 7 MT cigarettes (solid lines)

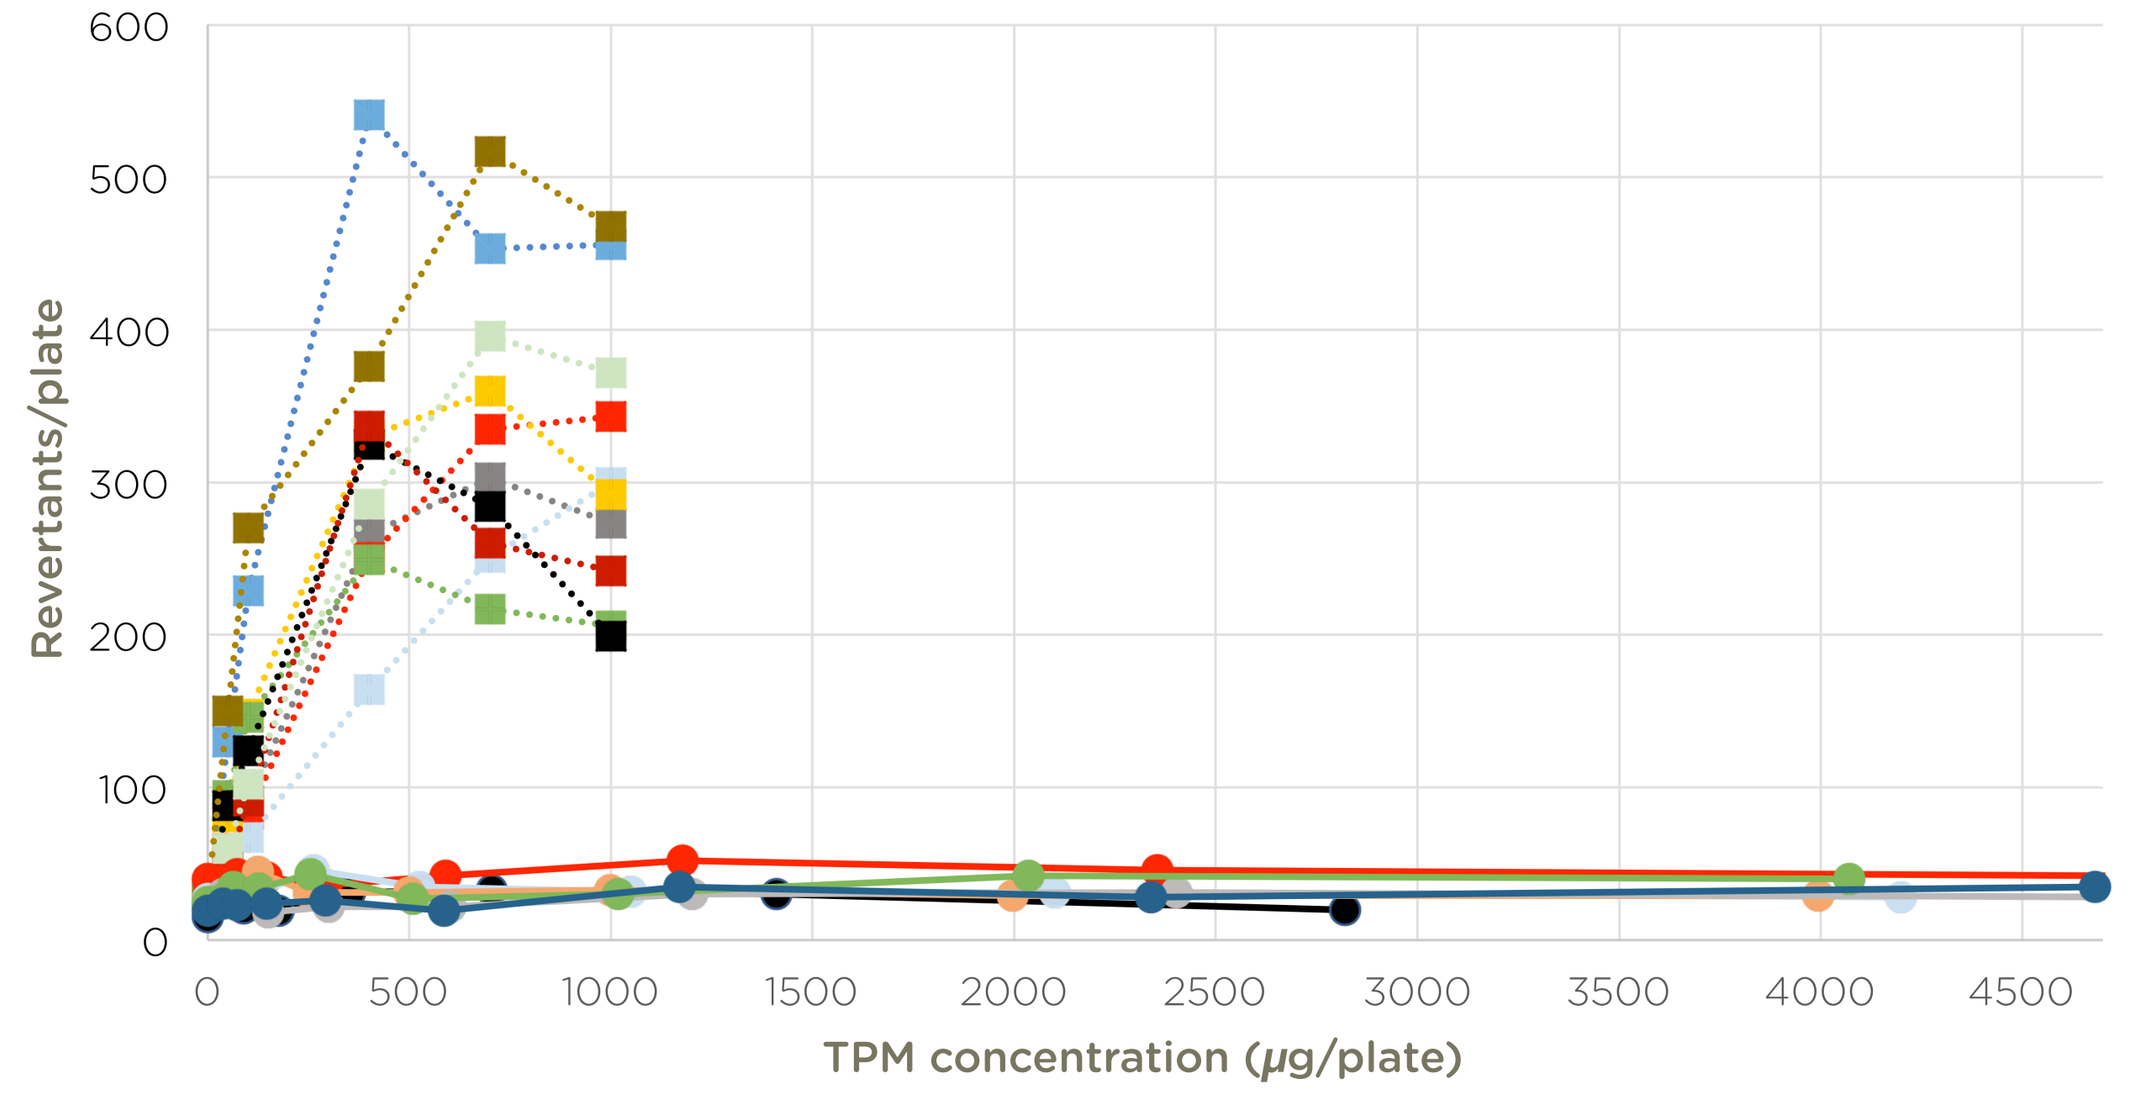
Revertants/plate
TPM concentration (µg/plate)
Cigarettes from DeMarini et al (2008):
2R4F Kentucky reference cigarette
Commercial US "low-nitrosamine tobacco" light cigarette
Experimental 100% reconstituted tobacco cigarette
Commercial US "low-ignition-propensity" cigarette
Experimental 100% Burley tobacco cigarette
Experimental 100% flue-cured cigarette
Commercial US ultra-low tar, non-menthol cigarette
Commercial US full-flavor, non-menthol cigarette
Commercial US light non-menthol cigarette prepared under FTC smoking regimen
Commercial US light non-menthol cigarette prepared under Massachusetts intense-smoking machine regimen

Figure 3–5. *In vitro* MN assay for combustible reference 3R4F (dotted line ■) and 7 MT e-condensates (solid lines). Top panel is cytotoxicity; bottom panel is the %MN.

Figure 3. 4-hr treatment without S9

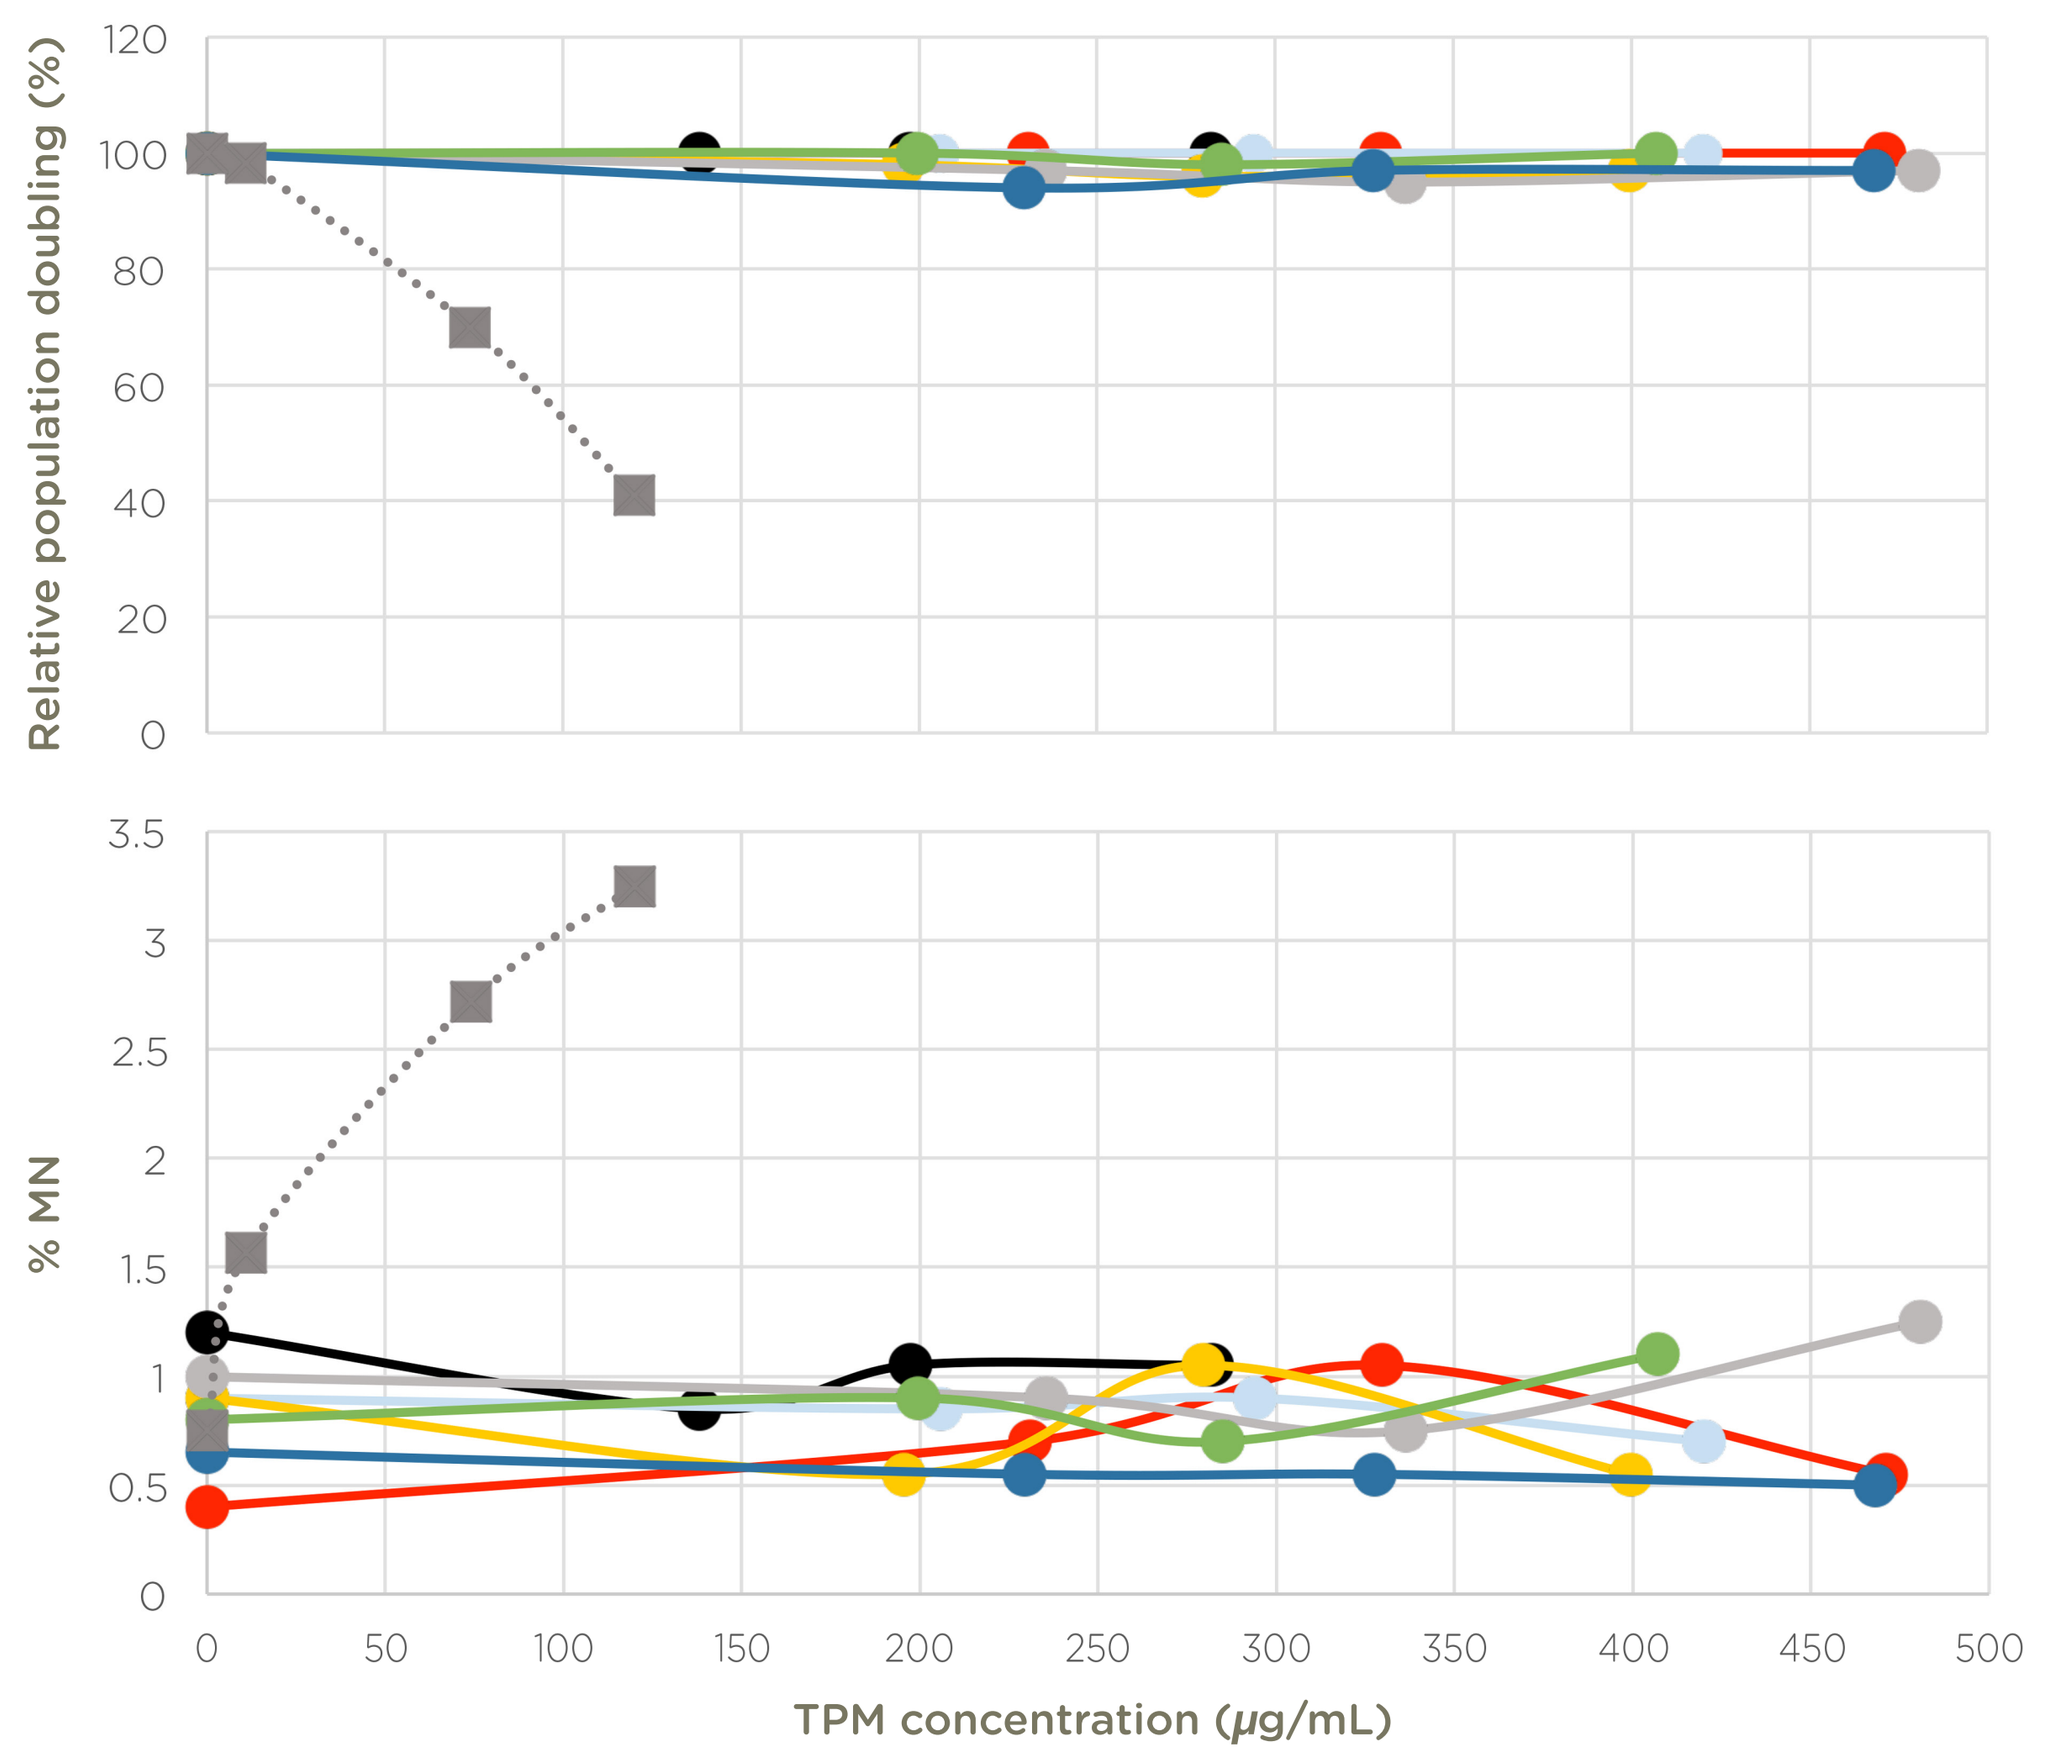
Relative population doubling (%)
% MN
TPM concentration (µg/mL)

Figure 4. 4-hr treatment with S9

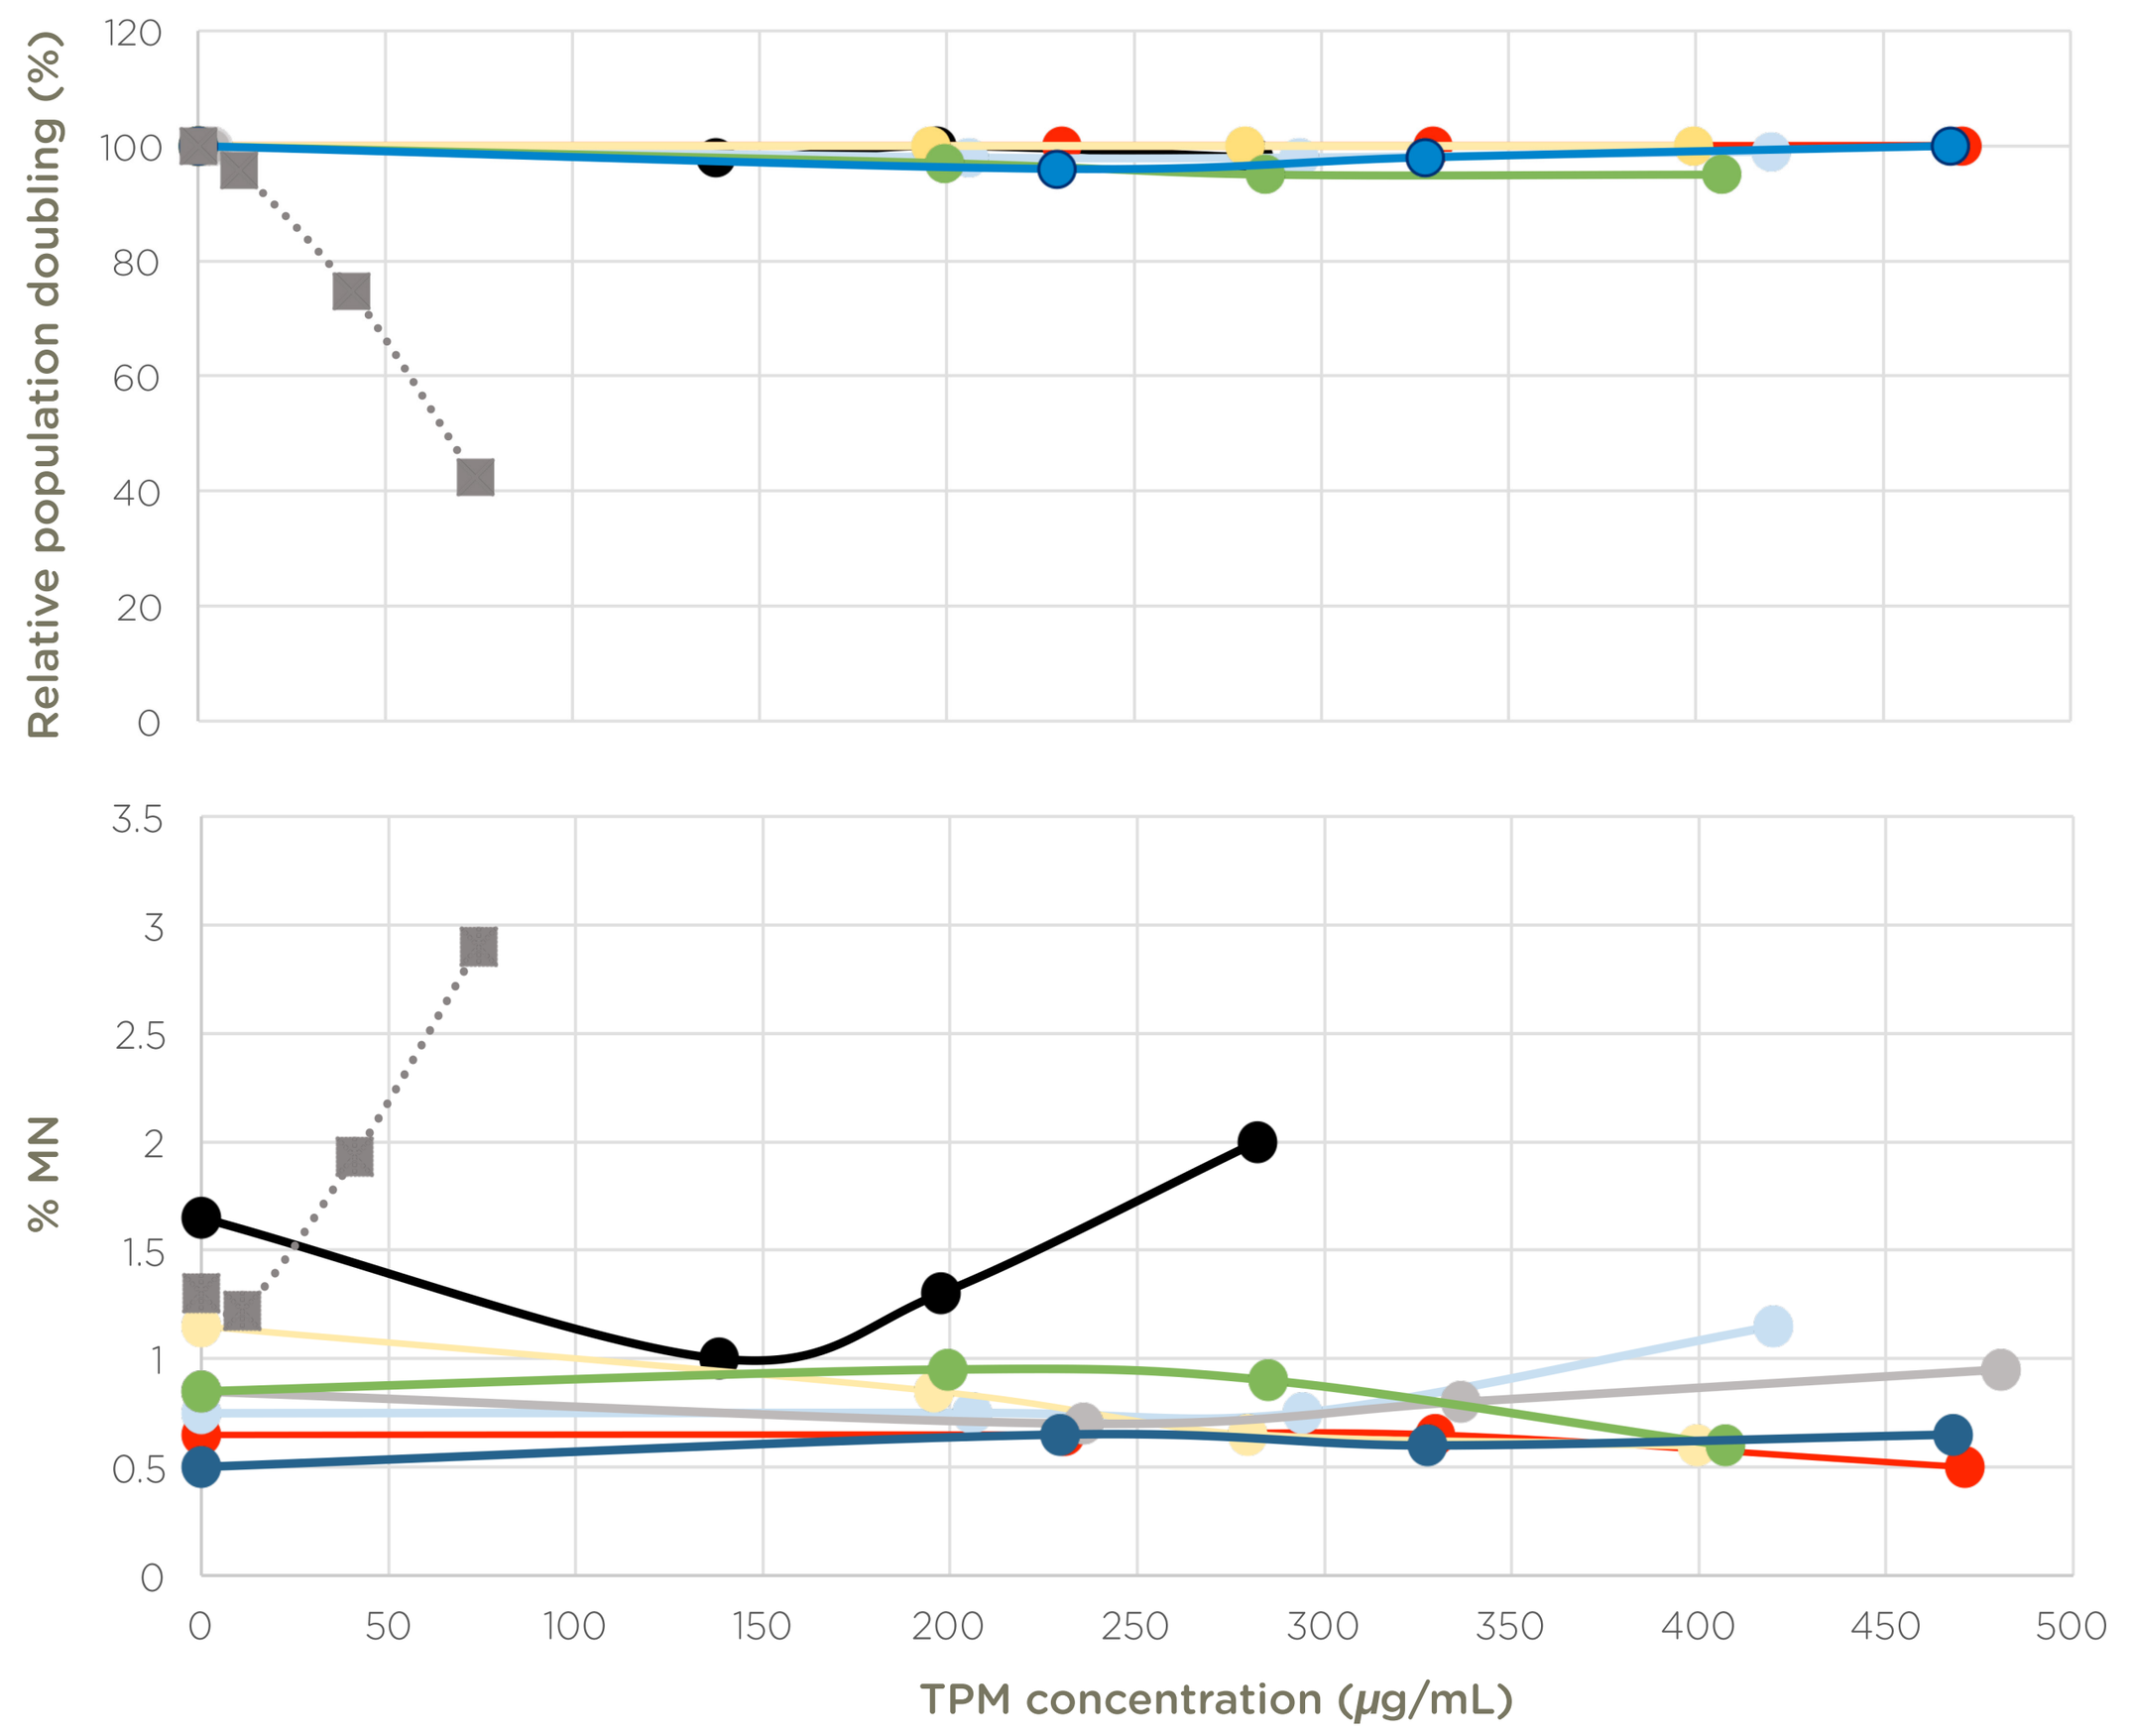
Relative population doubling (%)
% MN
TPM concentration (µg/mL)

Figure 5. 27-hr treatment without S9

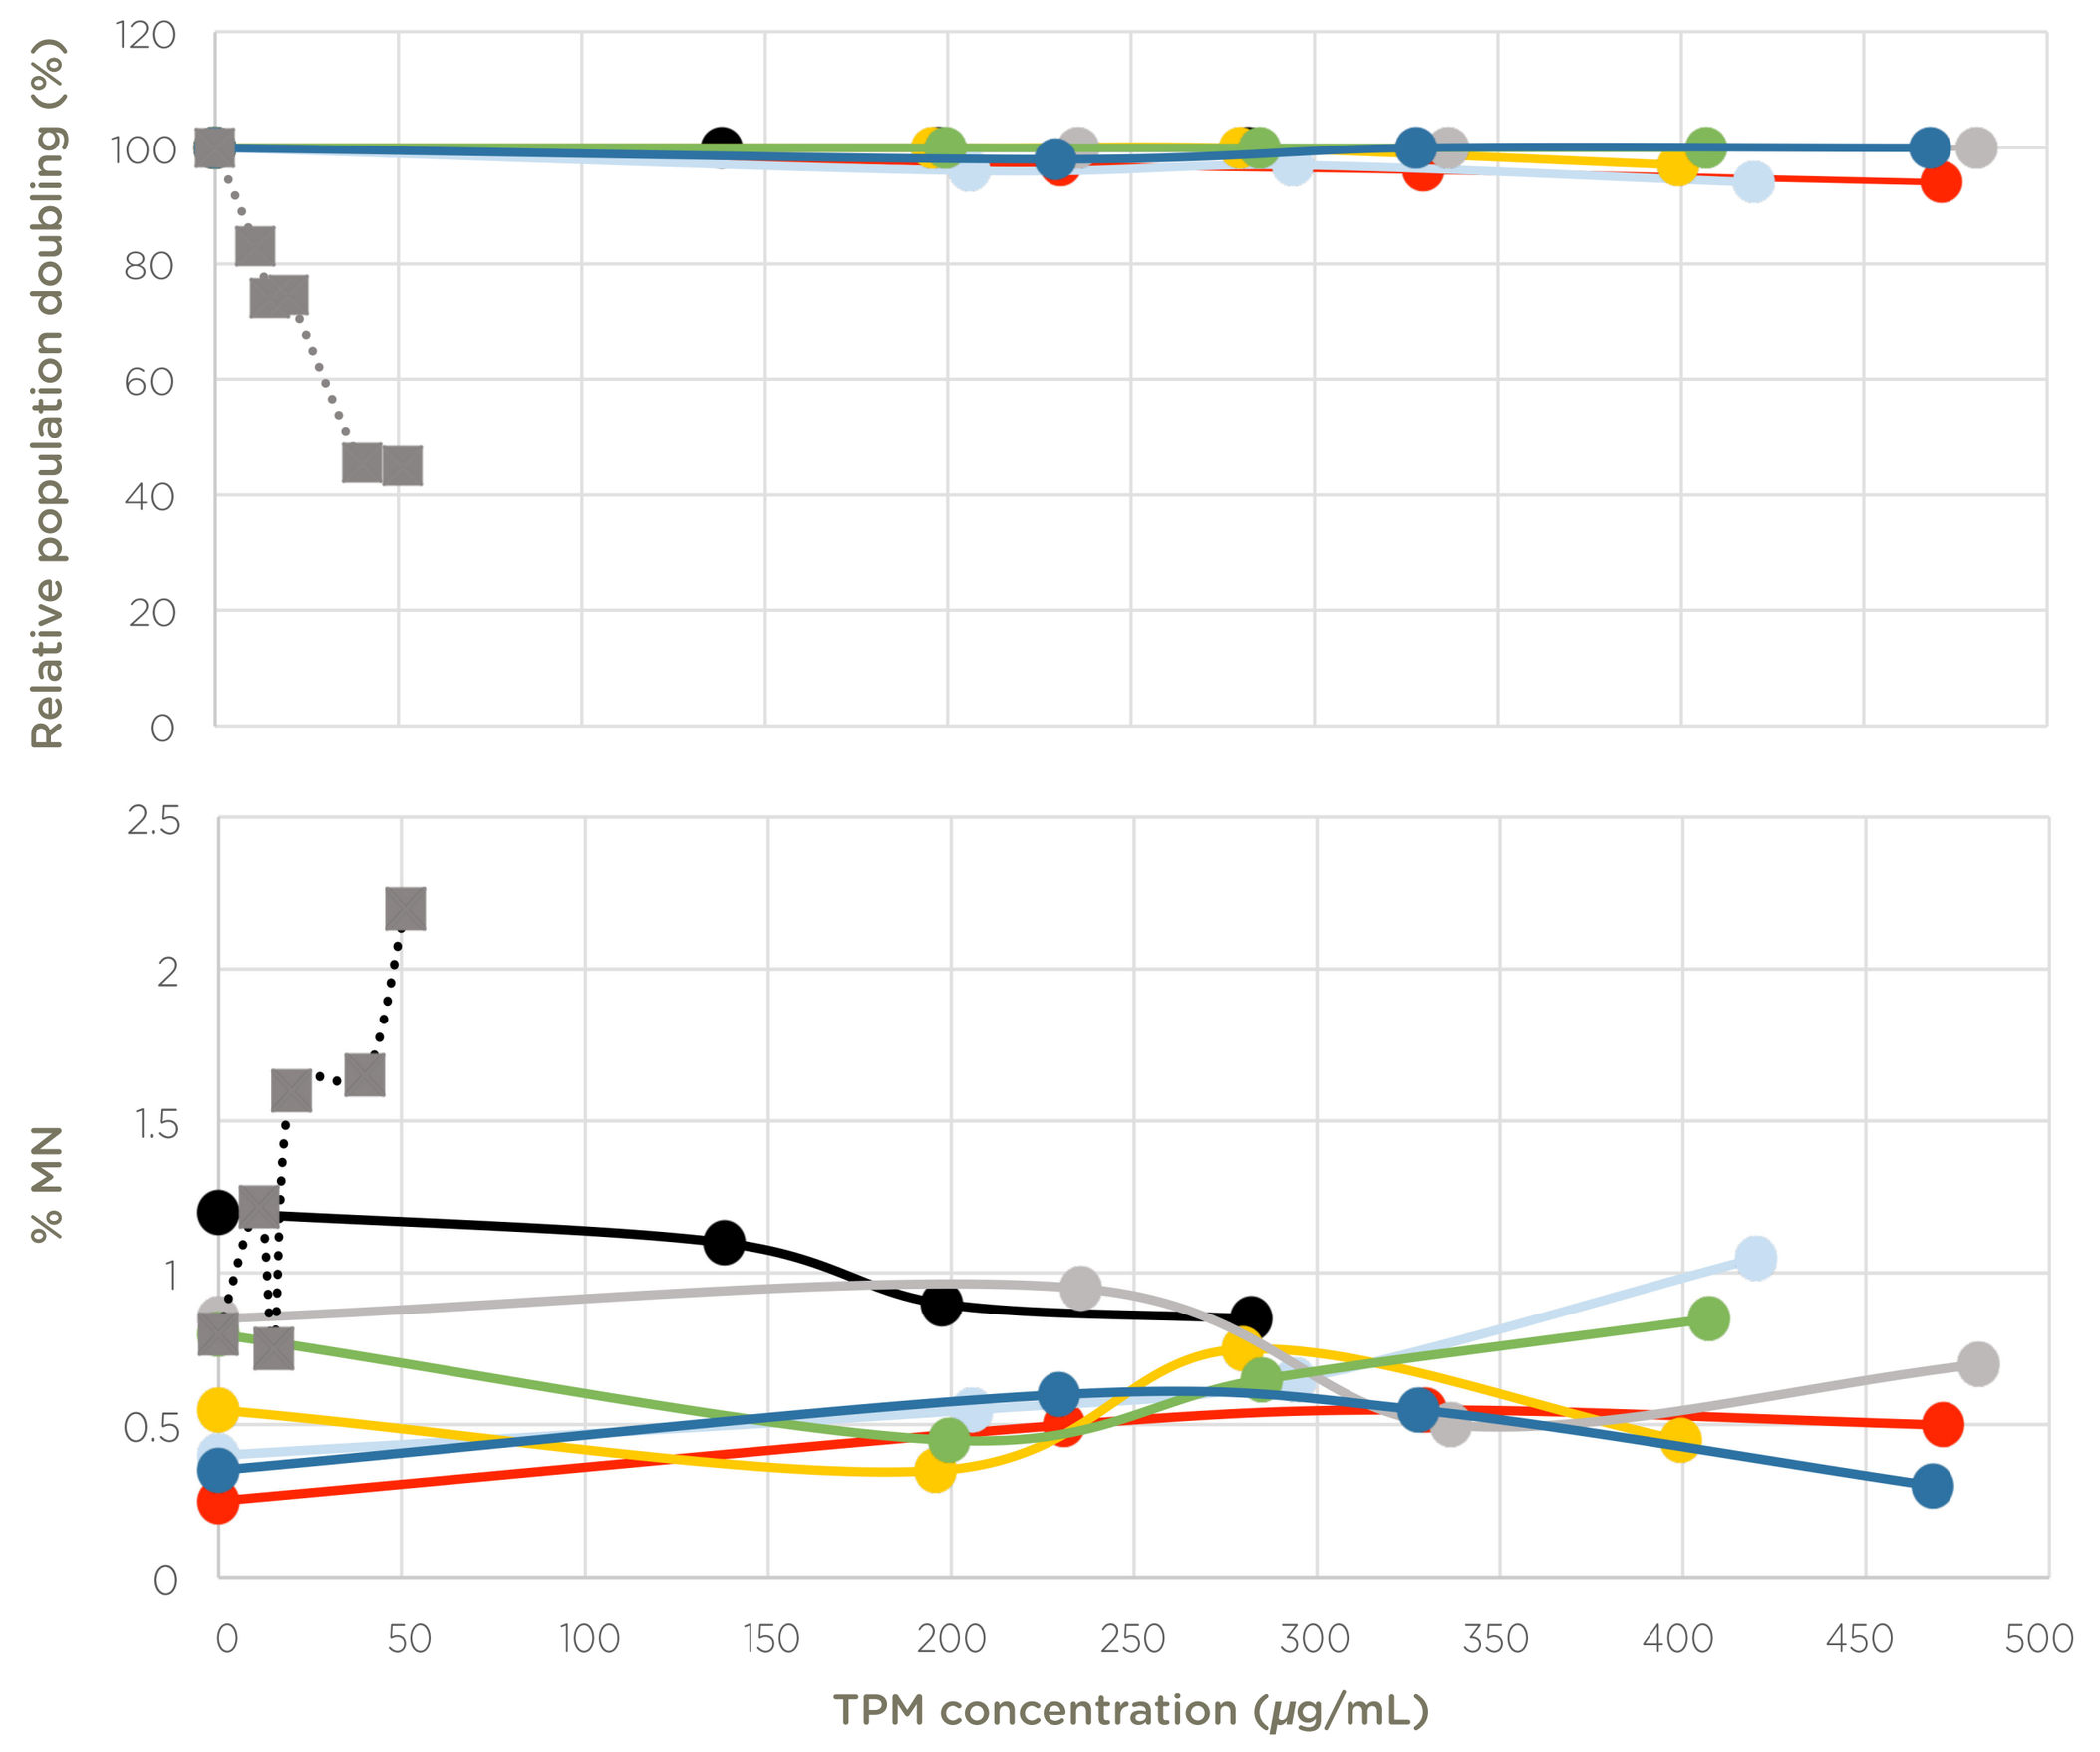
Relative population doubling (%)
% MN
TPM concentration (µg/mL)

Figure 7. Cytotoxicity (NRU assay) of BALB/c 3T3 cells exposed to 7 different MT e-cigarette condensates. The percent viability is expressed relative to the solvent control (ethanol)

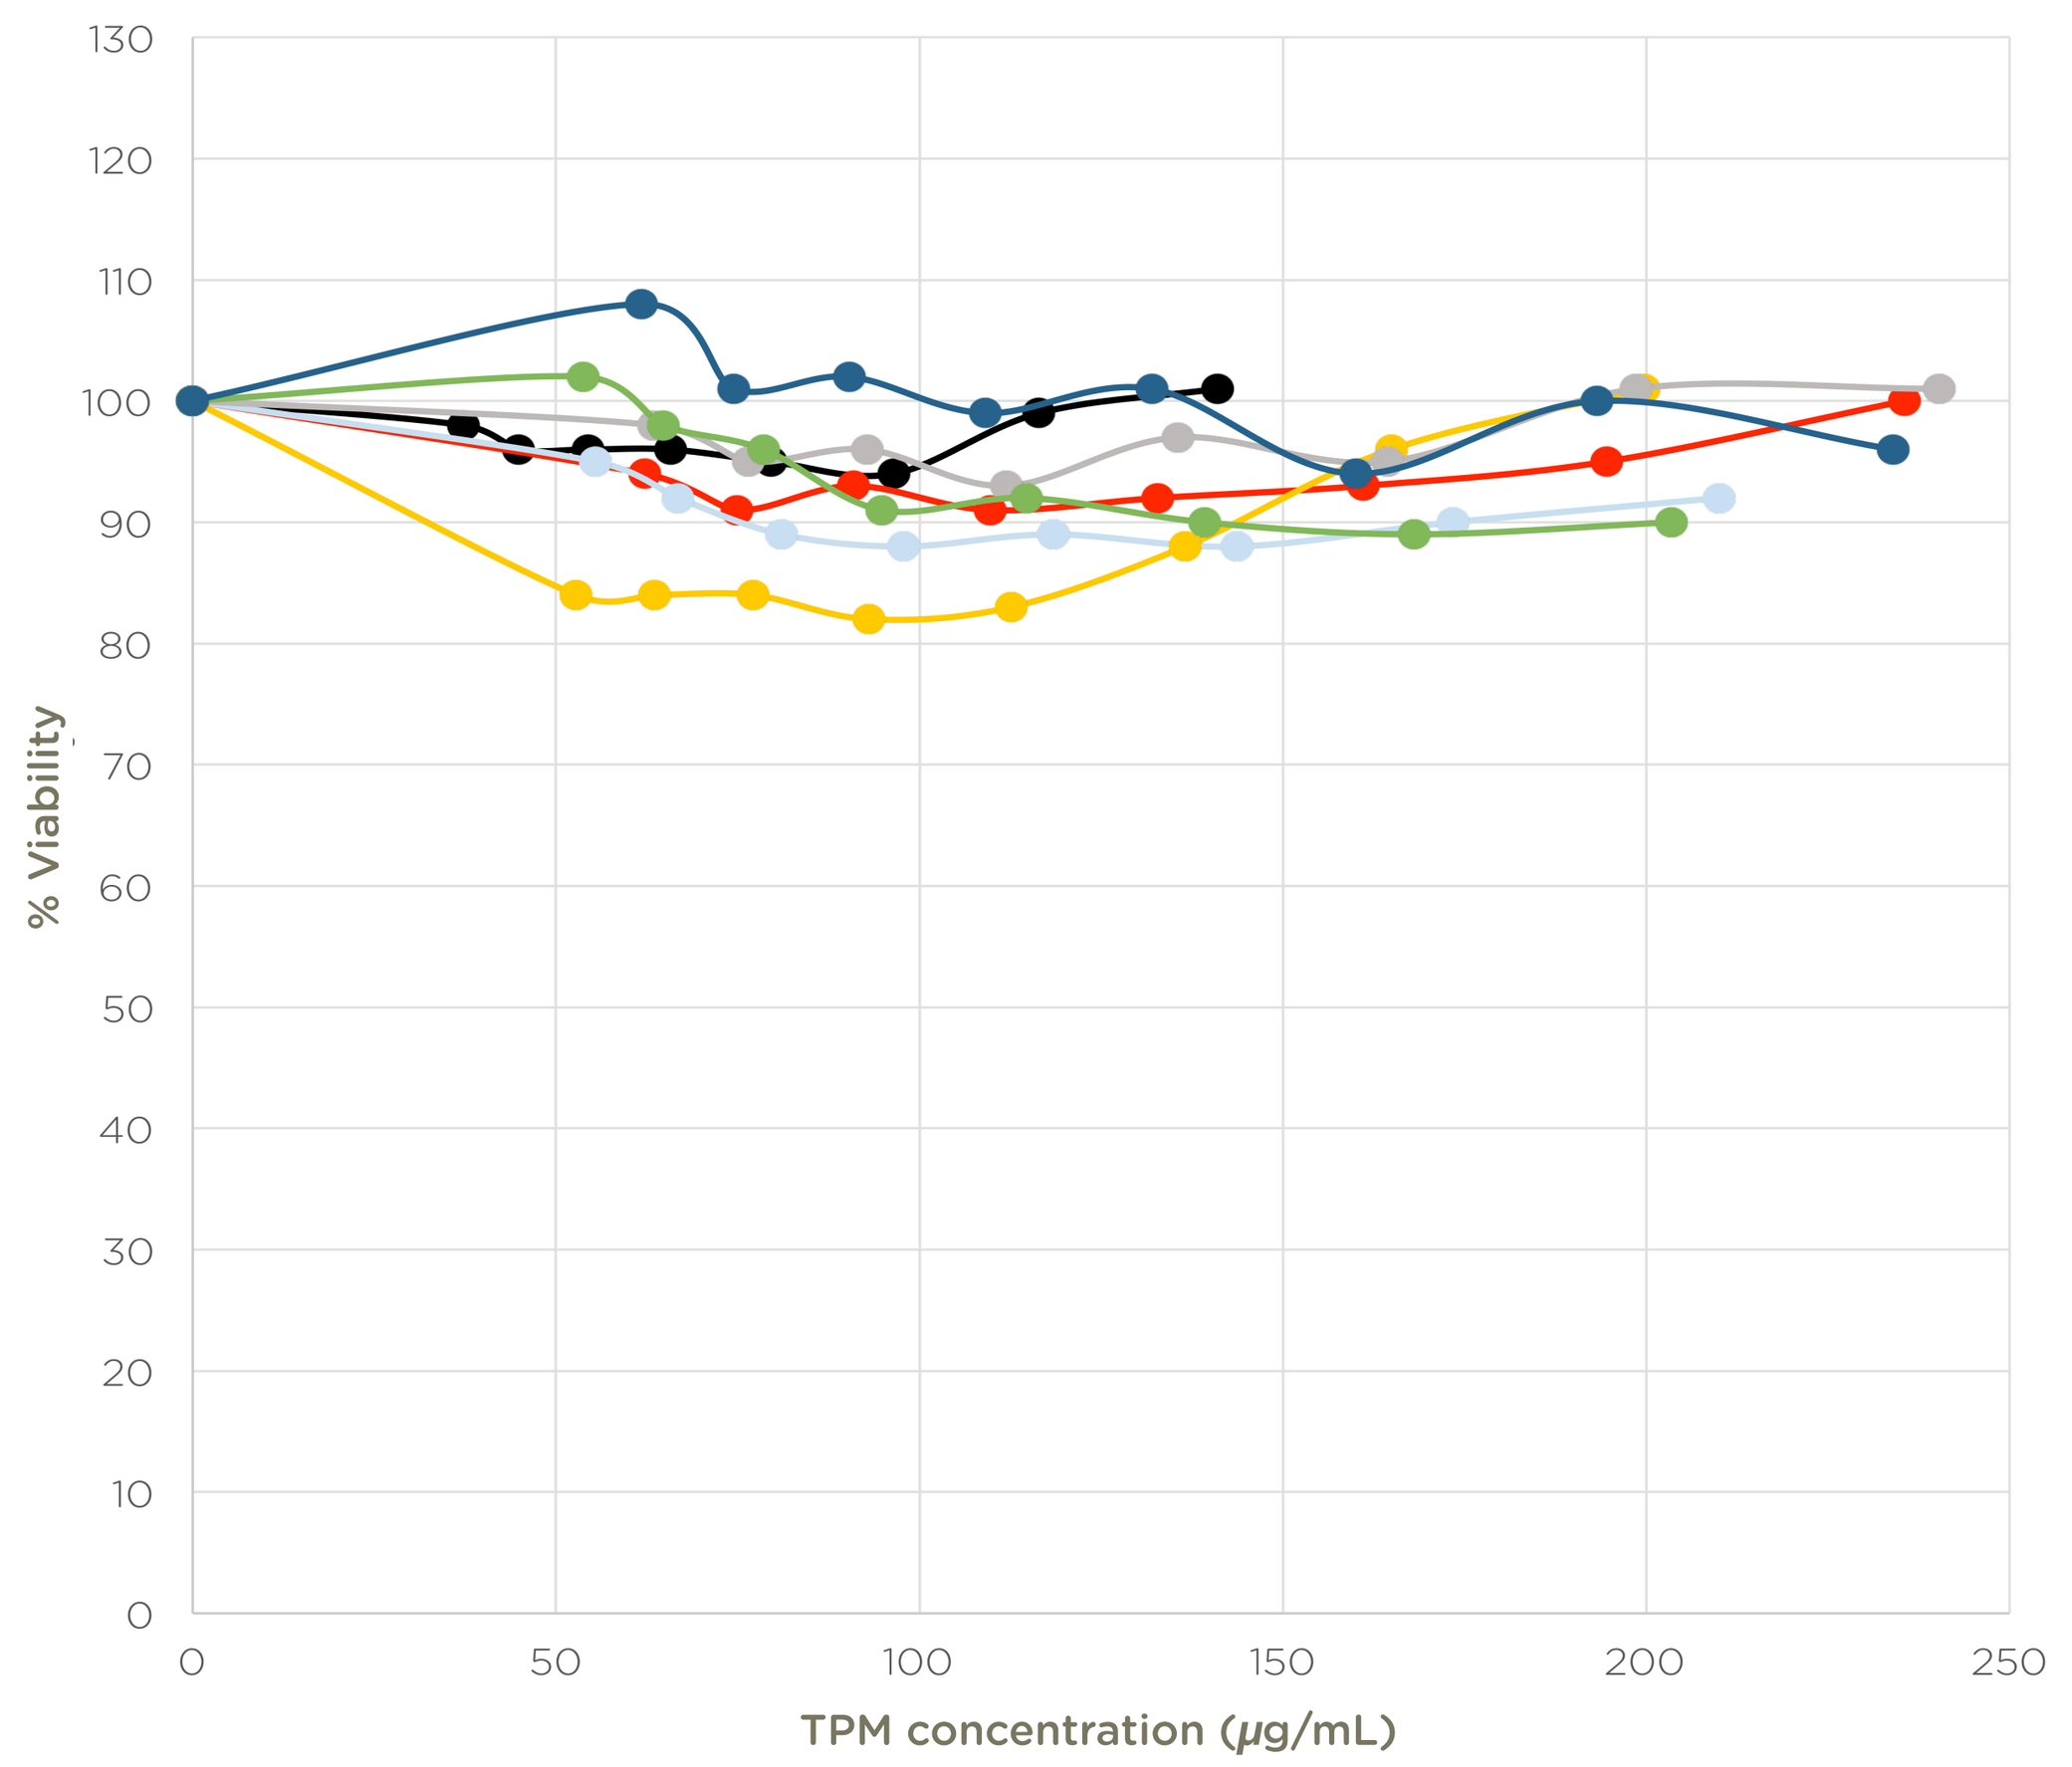
% Viability
TPM concentration (µg/mL)

e-Cigarettes: ● Carrier (50/50 mixture of PG/VG/5% nicotine) ● MT Bold Menthol ● MT Bold Classic ● MT Classic ● MT Menthol ● MT Summer Fusion ● MT Winter Mint