A Structure-Activity Relationship Based Approach to Tobacco Product Ingredient Hazard Assessment

Anderson, C; Awoyemi, O; Wei, Z; Zhang, J; Morgan, R; Lee, KM
Altria Client Services LLC, Richmond, VA 23219
Center for Research and Technology
Tobacco Science Research Conference 2022
September 11-14, 2022

Abstract

In silico tools allow for the rapid assessment of the potential toxicity of large groups of chemicals by associating specific chemical fragments with specific toxicological outcomes. These structure-activity relationship (SAR) tools have great potential to enhance pre-clinical ingredient and material hazard assessment while reducing time, cost, and animal use. These approaches are of particular interest in the effort to develop novel, reduced-harm tobacco products as they provide the basis for screening a variety of ingredients and materials that may have little to no experimental data in the literature. These developments are consistent with the FDA's commitment to advance new alternative methods (NAMs) in Predictive Toxicology in support of regulatory submissions.

In this case study, we selected a sample of flavoring ingredients used in Oral Tobacco Derived Nicotine (OTDN) products and evaluated through two commercially available *in silico* systems: 1) a statistical quantitative structure activity relationship (QSAR) system (CASE Ultra); 2) an expert rule-based system (DEREK). The chemical structure of each flavoring ingredient was input in simplified molecular-input line-entry system (SMILES) format and each system was independently queried for predictions focusing on genotoxicity, mutagenicity, and carcinogenicity. These predictions were evaluated for consensus between the two systems. The resulting consensus analysis assigned a concern level (I minor, II moderate, or III major) for each chemical. This NAMs approach provides an efficient strategy to identify ingredients of potential concern and guide future product development, research, and risk assessment.

Introduction

The past decade has seen the development of numerous novel, potentially reduced harm tobacco products, such as OTDN products. However, these diverse products often lack robust toxicity data, presenting a unique challenge to assess their relative risk compared to conventional products. Historically, these data gaps would have to be filled by *in vitro* and *in vivo* studies. A NAMS approach, such as *in silico* hazard assessment, may provide a rapid alternative that is compliant with the 3Rs principles of replacing, reducing, and refining the use of animals in research.

In this study, we have used two *in silico* structure activity (SAR) relationship-based systems to screen 267 ingredients used in OTDN products based on potentially hazardous structural features. We then evaluated the consensus of the output from these two systems and assigned a concern level to individual ingredients as part of the hazard assessment step of the risk assessment process.

Methods

Two *in silico* systems were used to evaluate mutagenicity, genotoxicity, and carcinogenicity: 1) a statistical QSAR system, CASE Ultra (MultiCASE, Inc.); 2) an expert rule-based SAR system, DEREK (Lhasa, Ltd.). The use of both a statistical and a rule-based system is considered best practice when performing an *in silico* hazard assessment (ICH, 2017, Hsu et al., 2018, Fitzpatrick et al., 2018, Hasselgren et al., 2019).

A total of 267 individual neat flavoring ingredients (e.g., oral ingredients generally recognized as safe (GRAS) by either the Flavor and Extract Manufacturers Association (FEMA) or the FDA and select ingredients used in commercial OTDN products) were selected for screening. The structure of each ingredients was input as a SMILES code. For CASE Ultra, modules GT1_BMUT, GT2_CHROM_CHL, GT2_CHROM_CHO, GT3_MNT_MOUSE,GT4_ML_ACT, GT4_ML_UNACT, CARC_MOUSE_F, CARC_MOUSE_M, CARC_RAT_F, and CARC_RAT_M were run. For DEREK, the endpoints mutagenicity, chromosome damage, and carcinogenicity were run for both bacterium and mammals.

Results were evaluated and binarized as either positive (any call higher than equivocal) or negative (equivocal or lower calls). The consensus between the two systems was compared for the three categories of endpoints (mutagenicity, genotoxicity, and carcinogenicity) to assign a concern level for each endpoint. 1. Concern Level I being no positive calls from either system = minor concern. 2. Concern Level II including a positive call from one system and a negative call from the other system = moderate concern. 3. Concern Level III consisting of positive calls from both systems = major concern.

Results

We screened a total of 267 flavoring ingredients considered for OTDN products. Of the total ingredients screened: 130 were Concern Level I for all categories, 98 were Concern Level II for at least one category, and 39 were Concern Level III for at least one category. Out of 206 total positive calls, most of the calls (107) were for genotoxicity, followed by carcinogenicity (81), with a relatively small number of calls for bacterial mutagenicity (18) (**Fig. 1**). A similar pattern was observed within the subset of 39 Concern Level III ingredients**,** with positive calls from both systems (21 genotoxicity, 14 carcinogenicity, four bacterial mutagenicity).

Table I provides an example of three flavoring ingredients along with their (Q)SAR calls and concern level classification. For example, cinnamaldehyde (CASRN 104-55-2) was assigned Concern Level III with respect to bacterial mutagenicity, indicating that the highest level of scrutiny should be paid to *in vitro* assays relevant to that endpoint. Cinnamaldehyde was assigned Concern Level II with respect to genotoxicity, indicating that a weight of evidence analysis of the literature is needed to determine whether the *in silico* call is supported by *in vivo* or *in vitro* studies. Cinnamaldehyde was assigned Concern Level I with respect to carcinogenicity, indicating that there is no known structural basis for suspecting that cinnamaldehyde is carcinogenic.

Figure I. Distribution of Positive Calls

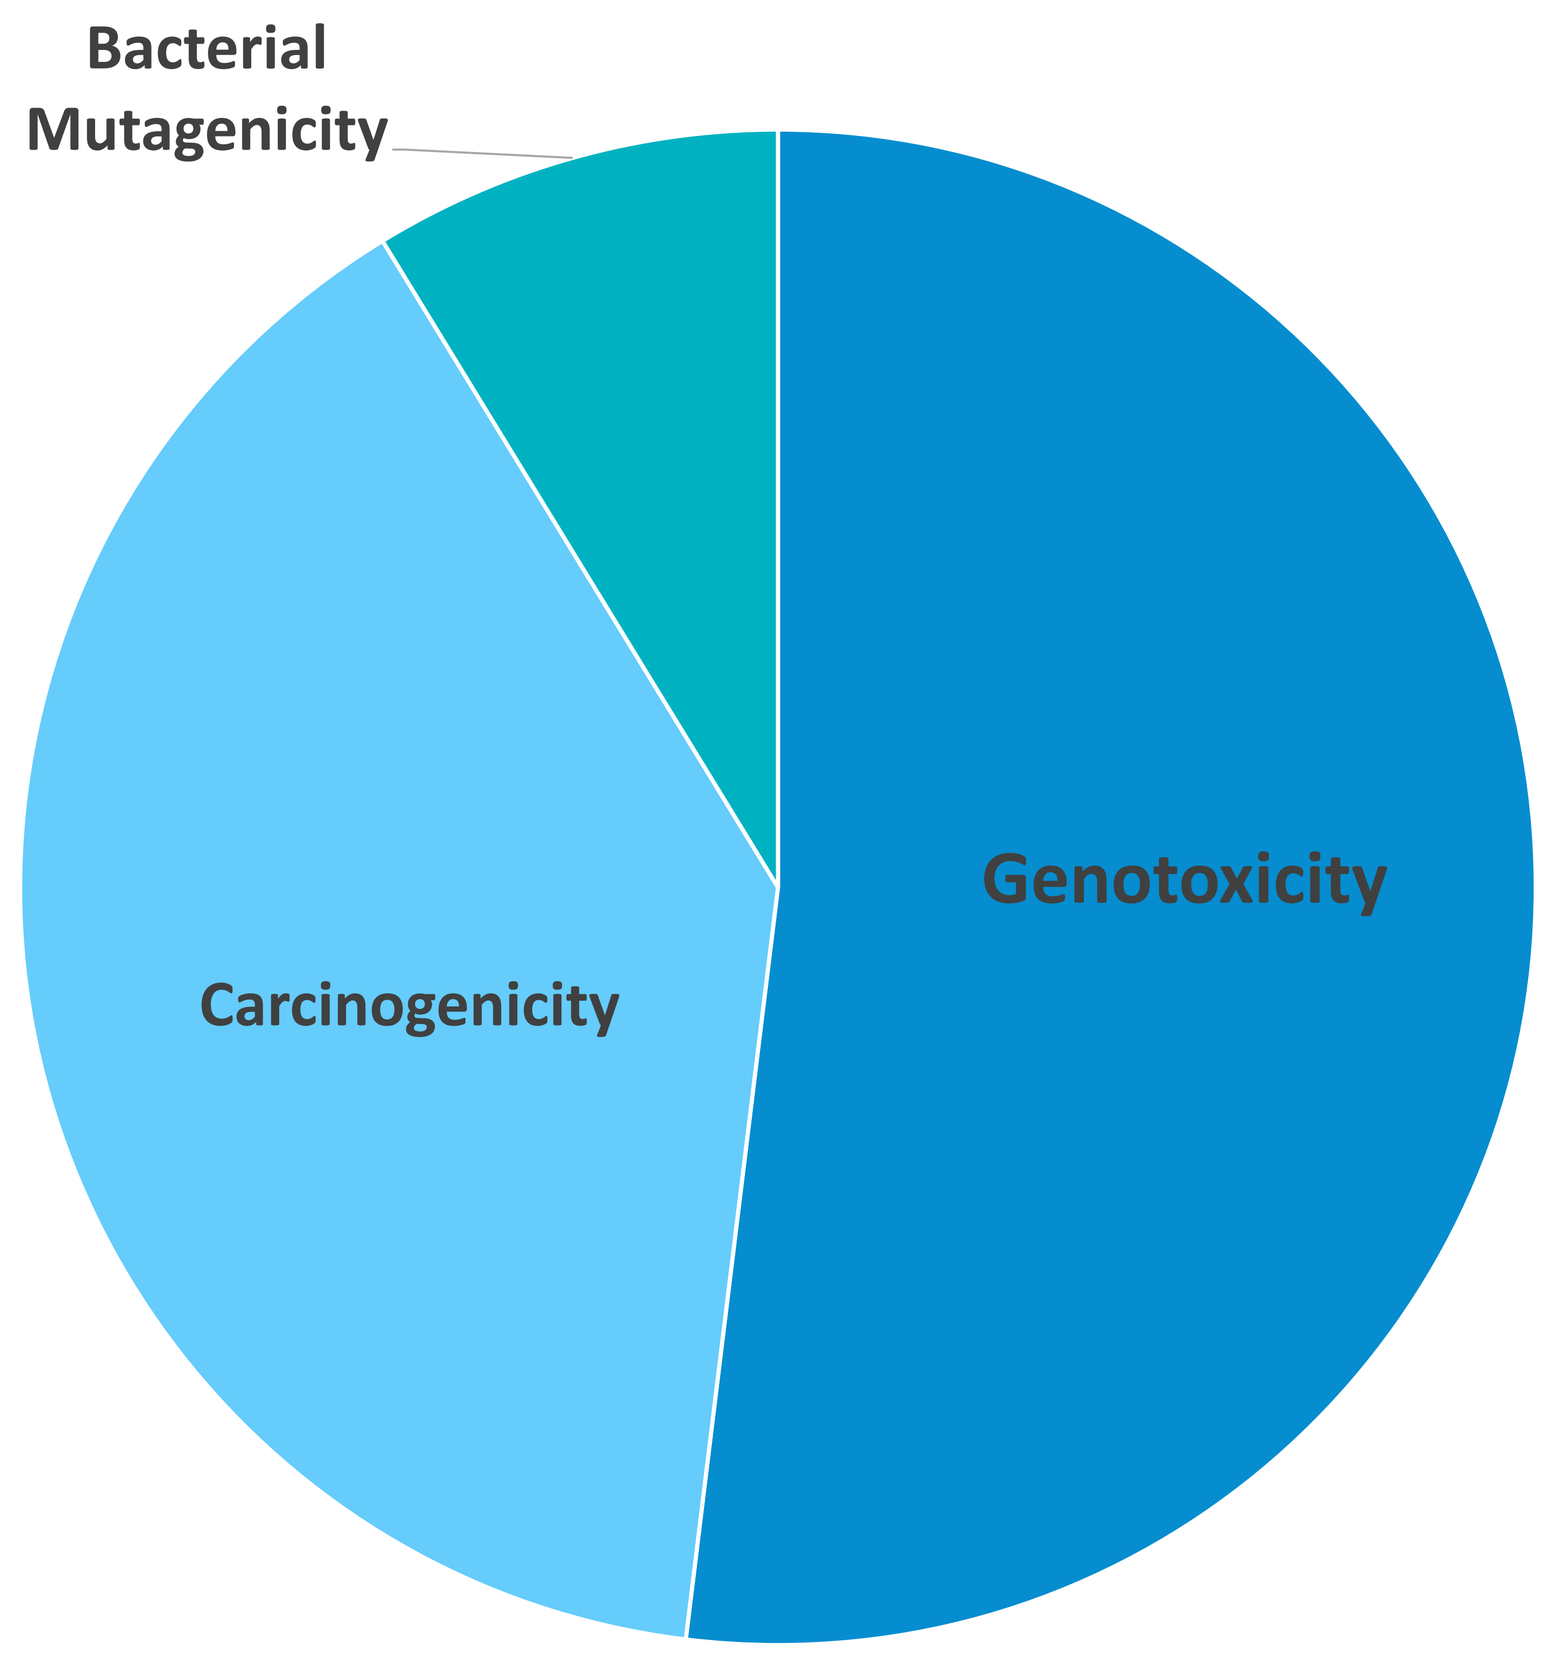
Bacterial Mutagenicity
Genotoxicity
Carcinogenicity

Conclusion

In silico hazard identification is an efficient strategy to screen and identify ingredients of concern and guide product development, research, and risk assessment. We demonstrated the ability to screen large sets of potential ingredients and efficiently categorize them into one of three concern levels based on the consensus output of two *in silico* hazard assessment systems. This approach allows for evidence-based resource prioritization driven by potentially hazardous structures in Concern Levels II & III ingredients. The specific nature of the alerts can serve to guide literature reviews, dose response assessment, and experimental design, as well as further analysis into metabolism and mixture effects.

We note that the hazard identification is only the first step of the risk assessment paradigm (hazard identification, dose response assessment, exposure assessment, risk characterization); this approach should be used as a starting point and prioritization tool. The specific determination of the suitability of any individual Concern Level II or III ingredient (such as cinnamaldehyde or linalool oxide, see Table 1) for inclusion in an OTDN product should be made based on the weight of evidence of a full risk assessment reflecting the use level intended in the product.

Table I. Example Ingredients, (Q)SAR calls, and Concern Levels

Ingredient Name	CASRN	SMILES	CASE Ultra	DEREK	Consensus
Acetoin	513-86-0	CC(O)C(C)=O	Bacterial Mutagenicity: (-) Genotoxicity: (-) Carcinogenicity (-)	Bacterial Mutagenicity: (-) Genotoxicity: (-) Carcinogenicity: (-)	Concern Level I Concern Level I Concern Level I
Cinnamaldehyde	104-55-2	O=CC=Cc1ccccc1	Bacterial Mutagenicity: (+) Genotoxicity: (+) Carcinogenicity: (-)	Bacterial Mutagenicity: (+) Genotoxicity: (-) Carcinogenicity: (-)	Concern Level III Concern Level II Concern Level I
Linalool Oxide	1365-19-1	CC(C)=CCCC(C)(O)C1CO1	Bacterial Mutagenicity: (+) Genotoxicity: (+) Carcinogenicity: (+)	Bacterial Mutagenicity: (+) Genotoxicity: (+) Carcinogenicity: (+)	Concern Level III Concern Level III Concern Level III

***In silico* hazard identification is an efficient strategy to identify ingredients of concern and guide product development, research, and risk assessment**

References

ICH (2017). ICH M7(R1) Assessment and Control of DNA Reactive (Mutagenic) Impurities in Pharmaceuticals to Limit Potential Carcinogenic Risk, ICH Harmonised Guideline
Hsu, Chia-Wen., Hewes, Kurt P., Stavitskaya, L., Kruhlak, Naomi L. (2018). Construction and application of (Q)SAR models to predict chemical-induced in vitro chromosome aberrations, Regulatory Toxicology and Pharmacology, 99, 274-288
Fitzpatrick JM, Roberts DW, Patlewicz G. (2018). An evaluation of selected (Q)SARs/expert systems for predicting skin sensitization potential. SAR QSAR Environmental Research, 29(6):439-468
Hasselgren, C., Ernst Ahlberg, Yumi Akahori, et al. (2019). Genetic toxicology in silico protocol, Regulatory Toxicology and Pharmacology, 107, 104403, https://doi.org/10.1016/j.yrtph.2019.104403

This scientific research is presented by Altria Client Services LLC (ALCS). ALCS affiliate companies are tobacco product manufacturers.

Altria Science